CHARTING A COMPLEX WORLD.

TECHATLAS

A DIVISION OF RACAPITAL

COVID-19 (CORONAVIRUS) THERAPEUTICS

UPDATED: FEBRUARY 11, 2022
Visit www.racap.com/covid-19 for latest version.
Anything wrong/outdated? Please contact covidteam@racap.com.
Strategic Overview & Instructions on How to Use This map

DISEASE SEVERITY AND PROGRESSION TIMELINE

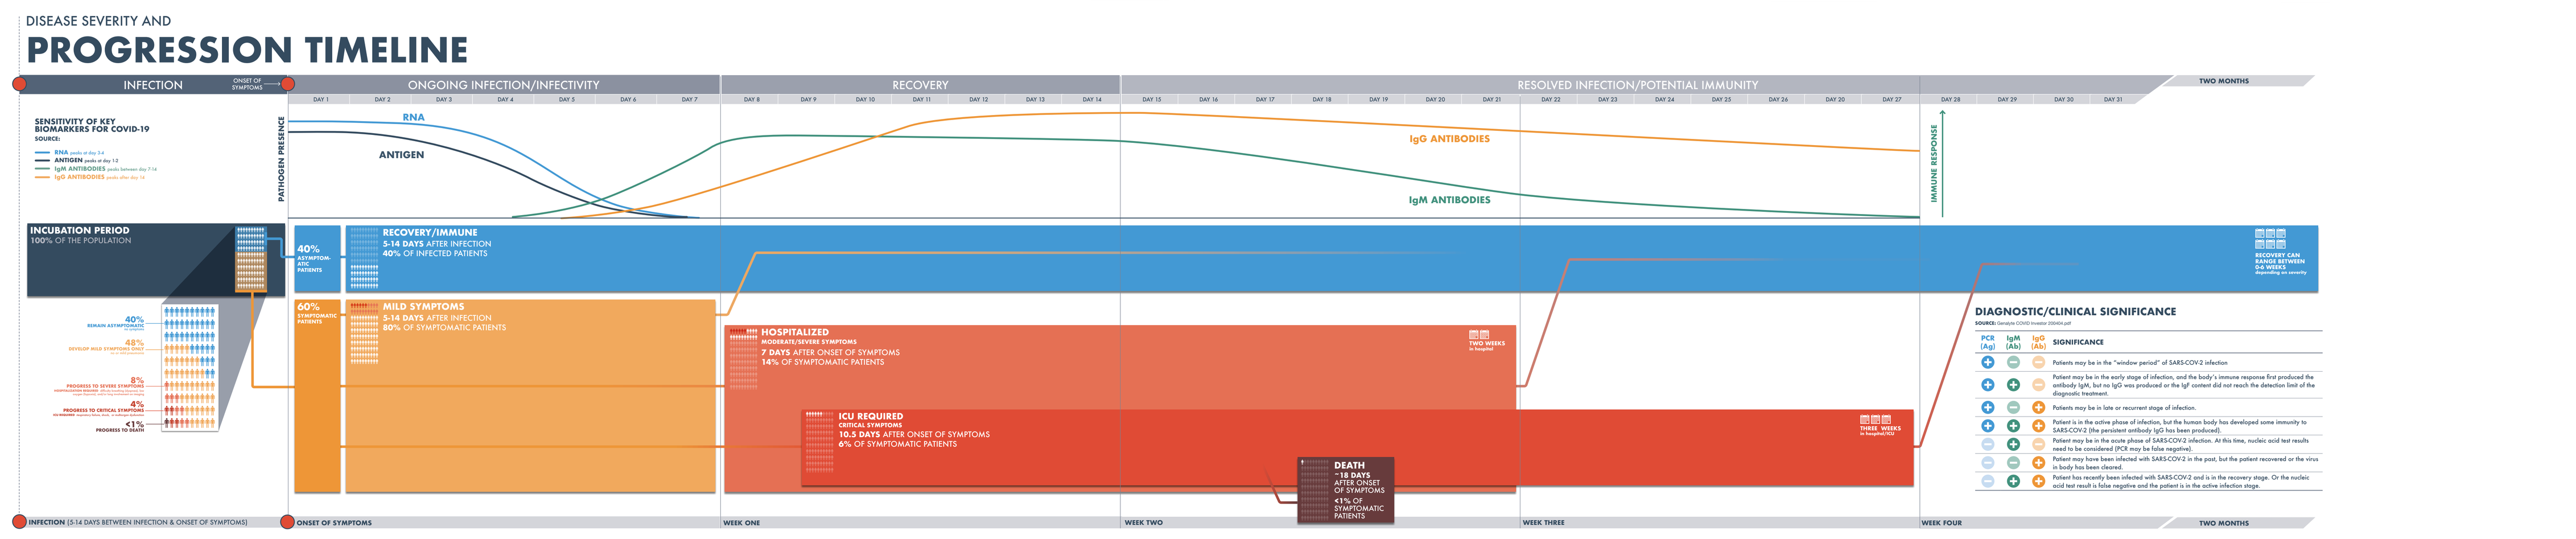

INFECTION
ONGOING INFECTION/INFECTIVITY
RECOVERY
RESOLVED INFECTION/POTENTIAL IMMUNITY
TWO MONTHS
SENSITIVITY OF KEY BIOMARKERS FOR COVID-19
RNA
ANTIGEN
IgG ANTIBODIES
IgM ANTIBODIES
PATHOGEN PRESENCE
IMMUNE RESPONSE
INCUBATION PERIOD
100% OF THE POPULATION
40% ASYMPTOMATIC PATIENTS
RECOVERY/IMMUNE
5-14 DAYS AFTER INFECTION
40% OF INFECTED PATIENTS
60% SYMPTOMATIC PATIENTS
MILD SYMPTOMS
5-14 DAYS AFTER INFECTION
80% OF SYMPTOMATIC PATIENTS
HOSPITALIZED
MODERATE/SEVERE SYMPTOMS
7 DAYS AFTER ONSET OF SYMPTOMS
14% OF SYMPTOMATIC PATIENTS
ICU REQUIRED
CRITICAL SYMPTOMS
10.5 DAYS AFTER ONSET OF SYMPTOMS
6% OF SYMPTOMATIC PATIENTS
DEATH
~18 DAYS AFTER ONSET OF SYMPTOMS
<1% OF SYMPTOMATIC PATIENTS
TWO WEEKS
THREE WEEKS
RECOVERY CAN RANGE BETWEEN 0-6 WEEKS
40% REMAIN ASYMPTOMATIC
48% DEVELOP MILD SYMPTOMS
8% PROGRESS TO SEVERE SYMPTOMS
4% PROGRESS TO CRITICAL SYMPTOMS
<1% PROGRESS TO DEATH
DIAGNOSTIC/CLINICAL SIGNIFICANCE
PCR (Ag)
IgM (Ab)
IgG (Ab)
SIGNIFICANCE
Patients may be in the "window period" of SARS-COV-2 infection
Patient may be in the early stage of infection, and the body's immune response first produced the antibody IgM, but no IgG was produced or the IgG content did not reach the detection limit of the diagnostic treatment.
Patients may be in late or recurrent stage of infection.
Patient is in the active phase of infection, but the human body has developed some immunity to SARS-COV-2 (the persistent antibody IgG has been produced).
Patient may be in the acute phase of SARS-COV-2 infection. At this time, nucleic acid test results need to be considered (PCR may be false negative).
Patient may have been infected with SARS-COV-2 in the past, but the patient recovered or the virus in body has been cleared.
Patient has recently been infected with SARS-COV-2 and is in the recovery stage. Or the nucleic acid test result is false negative and the patient is in the active infection stage.
INFECTION (5-14 DAYS BETWEEN INFECTION & ONSET OF SYMPTOMS)
ONSET OF SYMPTOMS
WEEK ONE
WEEK TWO
WEEK THREE
WEEK FOUR

MAP LEGEND

Preclinical Trial		ORAL	Oral delivery: could potentially be combined into an oral cocktail for outpatient/prophylactic use
Clinical Trial		SUBQ	Subcutaneous Delivery
Repurposed Therapeutic		IV	Intravenous Delivery
Prophylaxis Treatment		ID	Intradermal Delivery
Therapeutic Treatment Regardless of Hospitalization		INTRANASAL	Intranasal Delivery
Oral medication		INHALED	Inhaled Delivery
Treatment In Hospital Setting Only		IM	Intramuscular
Approved Therapeutic		RCT	Randomized Controlled Trial
Standard of Care		IST	Investigator-Sponsored Trial
EUA	Emergency Use Authorization	BARDA	In collaboration with BARDA
EFFECTIVE	Drug is effective	CEPI	In collaboration with CEPI
FAILED	Failed Trial		CHANGES HIGHLIGHTED IN YELLOW
DISCONTINUED	Discontinued Trial		CLICK TO SEE MAP UPDATE LOG >

This map is a compilation of the biotech industry's effort to rid the world of COVID-19.

Our visualization aims to convey not only individual innovations from specific companies but also the collective effort and sheer hustle of the entire industry.

The legacy of COVID-19 will hopefully include not only new treatments for an acute viral illness but also a greater appreciation of what society can accomplish when we unite against a common foe. We've already achieved remarkable innovations in many diseases: hepatitis C, HIV, melanoma, breast cancer, and cystic fibrosis, to name a few. What we need now is a sustained, collective effort to make medical advances available to everyone instead of overburdening patients with excessive out-of-pocket costs that discourage them from receiving appropriate care. This will require fundamentally reforming America's insurance system, building on the recent patches put in place after payers initially refused to cover COVID-19 testing and the uninsured were overwhelmed by the cost of hospitalization.

The Biotech Social Contract, as described in **The Great American Drug Deal**, affirms that we do not have to choose between innovation and affordability. We as a society can rise to the challenge of tackling all human diseases just as we have COVID-19, making investments that will permanently upgrade our health and that of future generations.

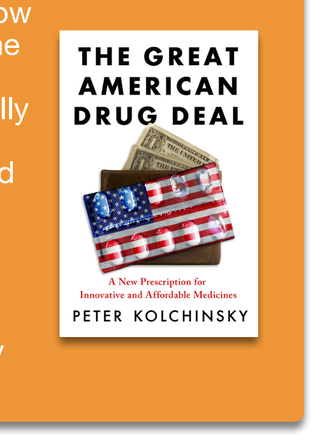

THE GREAT AMERICAN DRUG DEAL
A New Prescription for Innovative and Affordable Medicines
PETER KOLCHINSKY

COVID-19 THERAPEUTICS

To save lives, we need vaccines to prevent infection and drugs to treat people who become infected. Fortunately, many of both are in development, though we don't yet know what works. The first vaccines are likely to become available towards the end of 2020 for front-line workers and for masses in 1H21.

Drugs to treat infections may already be available if existing ones we have for other diseases turn out to work for COVID-19 (e.g. remdesivir, Actemra), but if we do need new ones (as we likely will), then those probably won't be available until September 2020 or later (need to run tests and manufacture).

COVID-19 DISEASE OVERVIEW

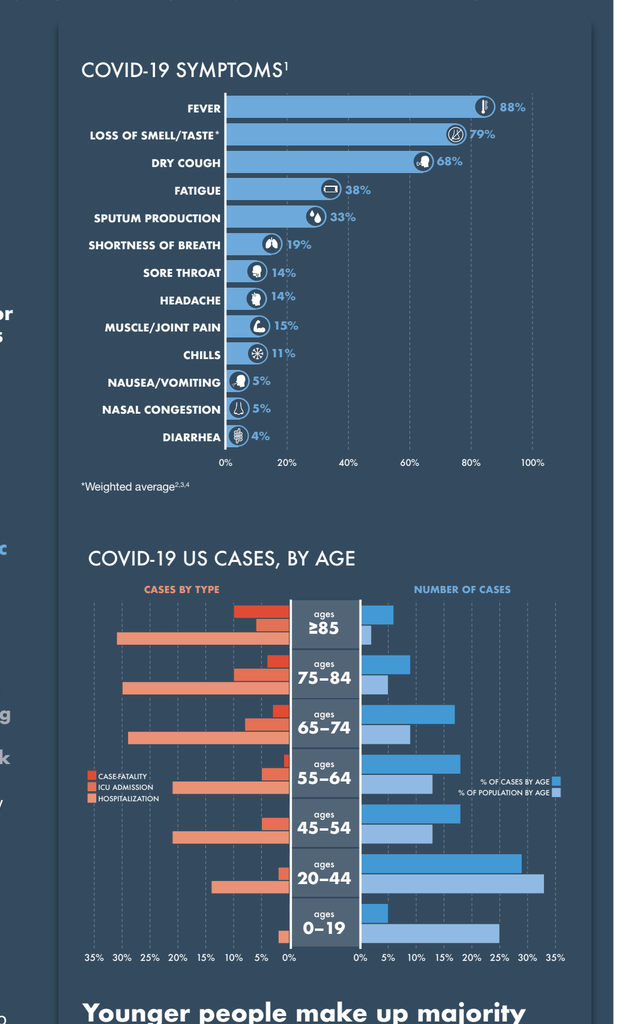

COVID-19 SYMPTOMS[1]
FEVER 88%
LOSS OF SMELL/TASTE* 70%
DRY COUGH 68%
FATIGUE 38%
SPUTUM PRODUCTION 33%
SHORTNESS OF BREATH 19%
SORE THROAT 14%
HEADACHE 14%
MUSCLE/JOINT PAIN 15%
CHILLS 11%
NAUSEA/VOMITING 5%
NASAL CONGESTION 5%
DIARRHEA 4%
*Weighted average[2,3]
COVID-19 US CASES, BY AGE
≥85
75–84
65–74
55–64
45–54
20–44
0–19
Younger people make up majority of the cases, while older people disproportionately get serious covid-19 and die.

SYMPTOMS:

Fever, dry cough, fatigue, and shortness of breath are the major symptoms associated with COVID-19. The CDC also includes loss of taste or smell, chills, muscle pain, headache, and sore throat as symptoms to watch for. While other symptoms have been reported, they occur much less frequently.

DISEASE PROGRESSION:

PRE-INFECTION

COVID-19 can be prevented through the use of basic precautions and (in the future) an effective vaccine.

POST-EXPOSURE

After exposure to SARS-CoV-2, it takes several days for symptoms to develop. During this incubation period, the infected person is able to transmit the virus despite being unaware of their illness. This is why basic protective measures like social distancing, hand washing, and mask wearing are critical. Most people develop symptoms between 2-14 days post-infection, with the majority developing symptoms by 5 days[5].

If we had a therapy that was safe, efficacious, and easy to administer, it could be given to people who have come into contact with infected individuals (such as caretakers or hospital workers) to try to prevent them from becoming infected. This kind of treatment is called post-exposure prophylaxis, and while there are currently many ongoing trials in this space, no medications are yet approved for this purpose. Those who believe they have been exposed to the virus should self-quarantine for 14 days to prevent spreading it to others.

MILD

Most patients who develop COVID-19 have a mild disease course and are able to manage their symptoms at home using over-the-counter medications that relieve pain and/or reduce fever (e.g., Tylenol/acetaminophen or Advil/ibuprofen). If you are diagnosed with COVID-19, please follow the advice of your healthcare professional.

Some "asymptomatic" patients with COVID-19 may not develop any noticeable symptoms and may not realize they have been infected. Because these individuals can still spread the virus, everyone should employ basic protective measures like social distancing, hand washing, and mask wearing (obviously none of those are necessary if you live by yourself and never see anyone else, though washing hands is still nice).

HOSPITALIZATION

Some patients who develop COVID-19 symptoms become seriously ill (1 out of every 6 diagnosed, but a lower rate amongst all, including undiagnosed, who are infected), develop difficulty breathing, and must be admitted to the hospital to manage these symptoms. Older people (≥65 years old) and those with other medical issues are most at risk of developing serious illness. Some identified risk factors include cardiovascular disease, diabetes, high blood pressure (hypertension), chronic lung disease, cancer, and chronic kidney disease (CKD). Typically, hospitalized patients have difficulty breathing and decreased levels of oxygen in their blood, requiring administration of supplemental oxygen[6,7].

Acute respiratory distress syndrome (ARDS) is a serious complication in patients with severe disease and is associated with a high risk of respiratory failure. These patients must be carefully managed in the intensive care unit, often with the use of mechanical ventilation, and have the highest risk of death from COVID-19. ARDS is common for hospitalized patients with COVID-19 (20-42%) and initial labored breathing can quickly progress to ARDS and ICU admission. In addition to lung injury, ARDS is also associated with kidney, liver, and heart complications, which can also increase the risk of mortality[8,9,10].

COVID-19 THERAPEUTICS: EFFICACY

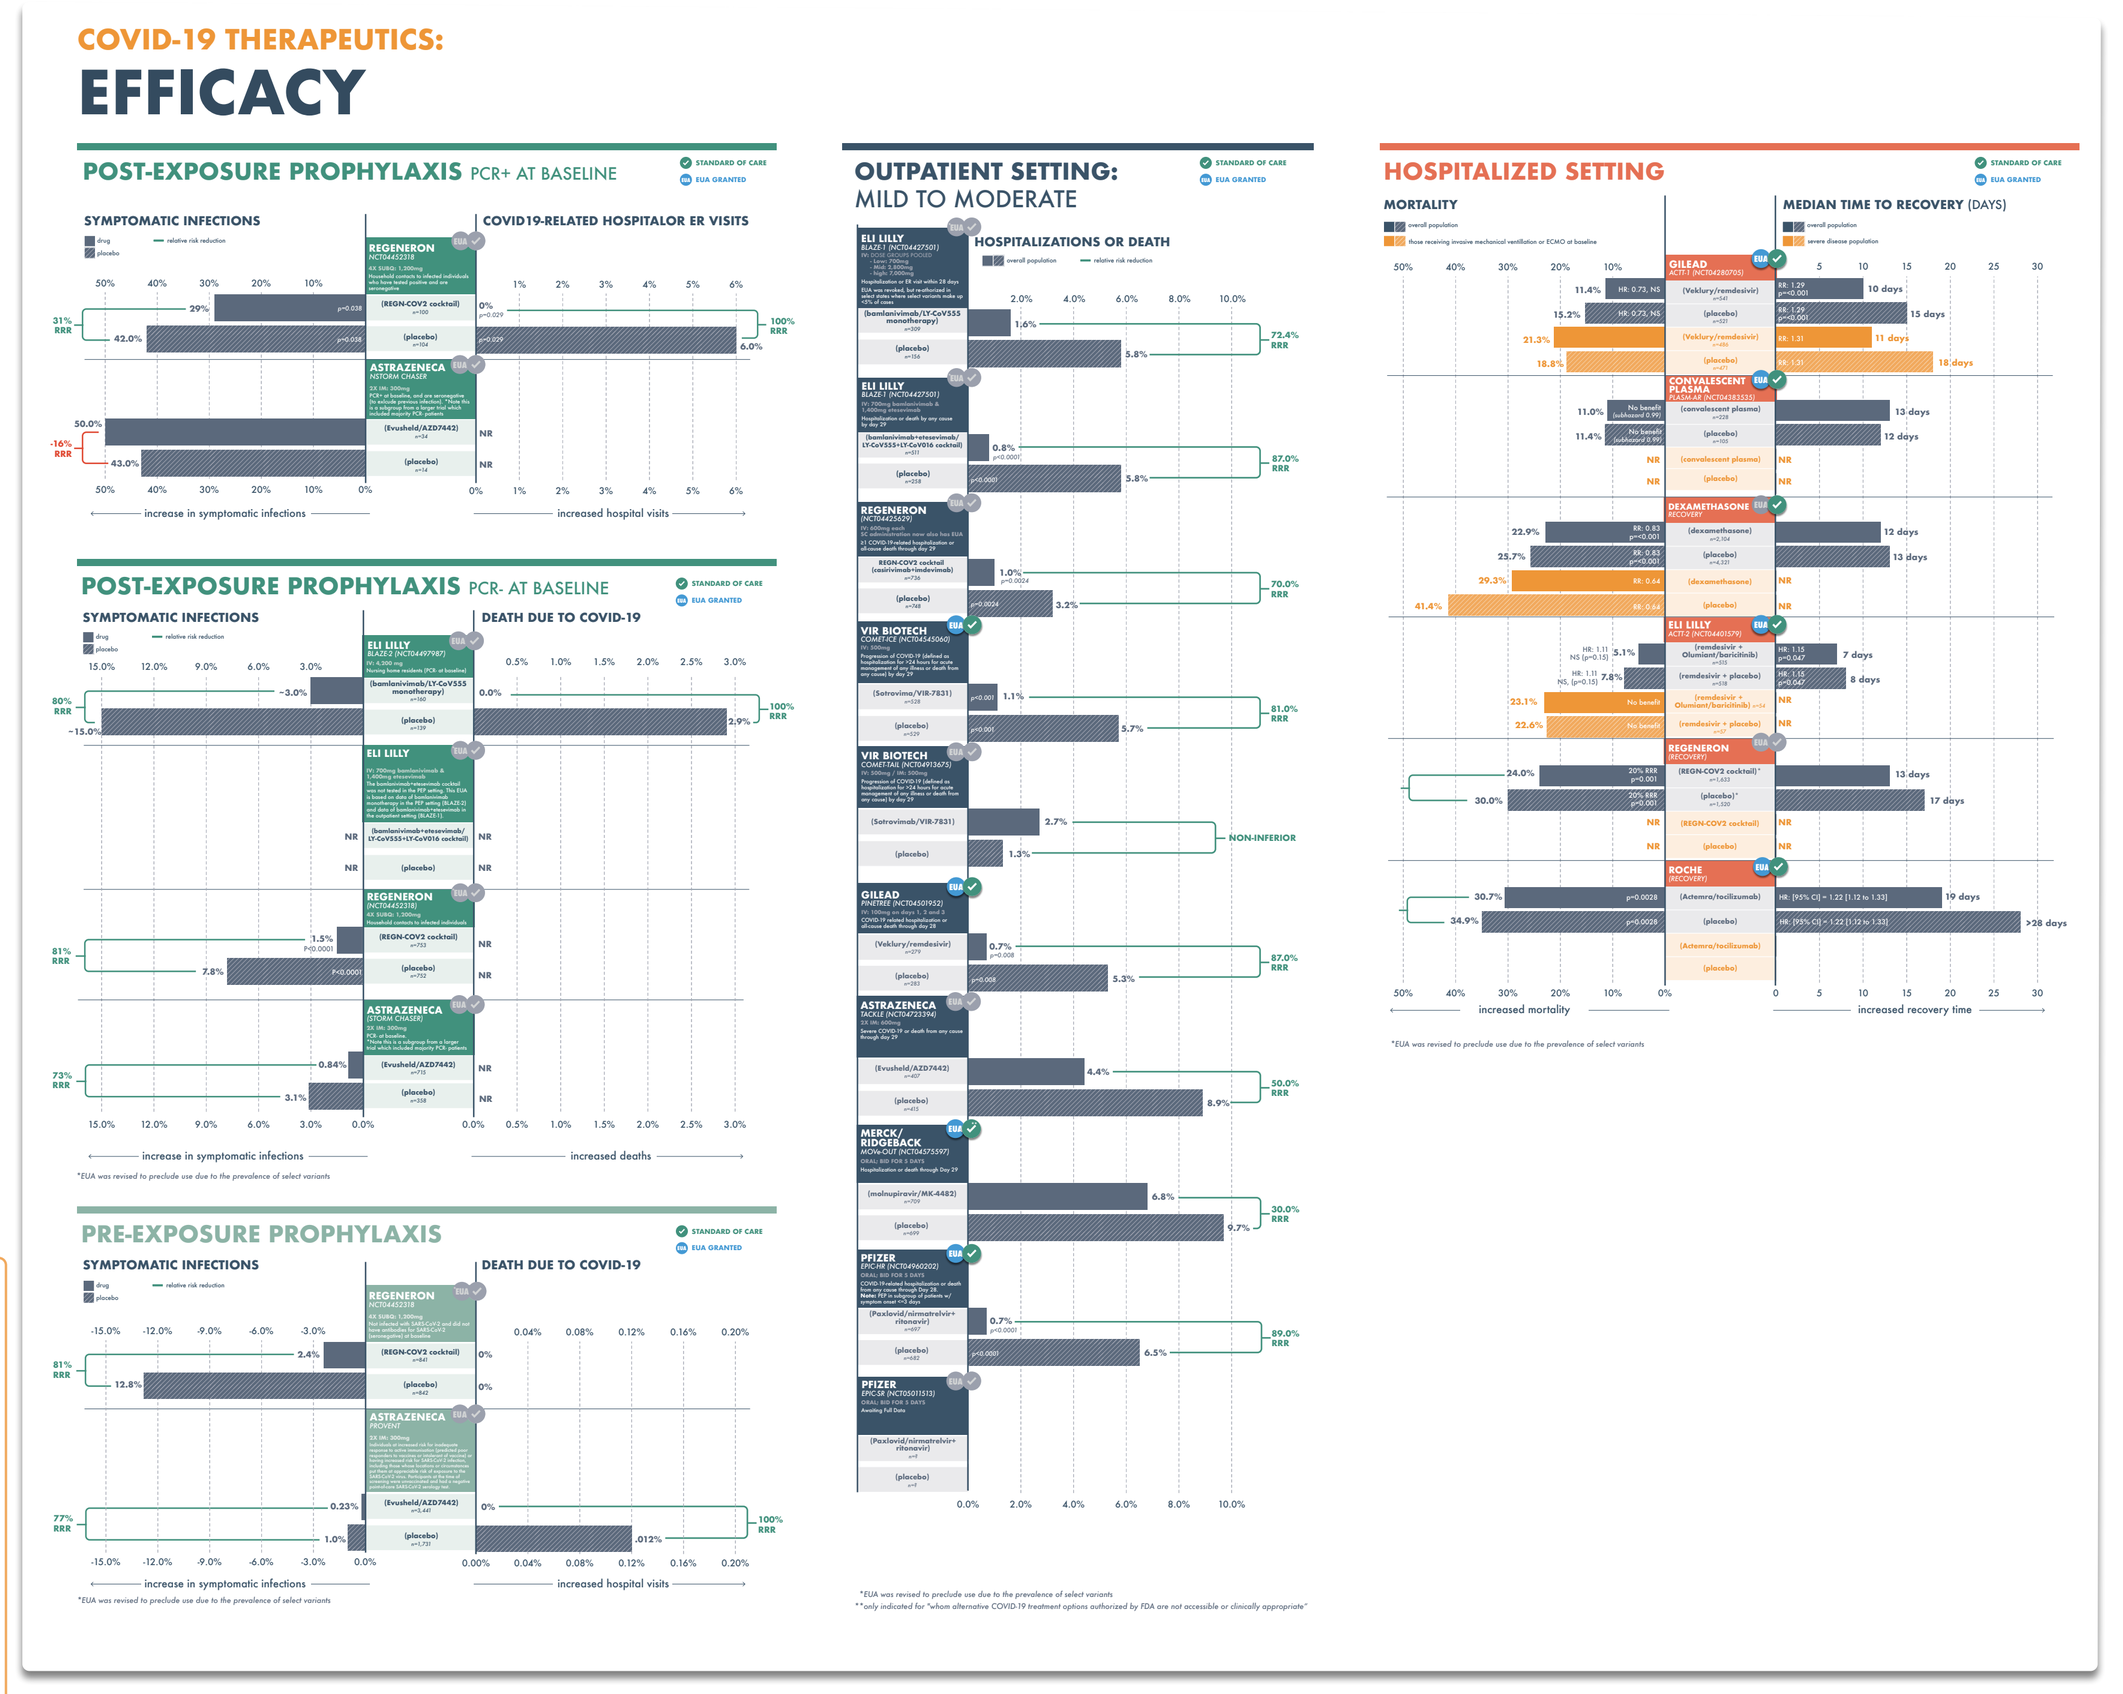

POST-EXPOSURE PROPHYLAXIS PCR+ AT BASELINE
SYMPTOMATIC INFECTIONS
COVID-19-RELATED HOSPITAL OR ER VISITS
REGENERON
ASTRAZENECA
POST-EXPOSURE PROPHYLAXIS PCR- AT BASELINE
DEATH DUE TO COVID-19
ELI LILLY
PRE-EXPOSURE PROPHYLAXIS
OUTPATIENT SETTING: MILD TO MODERATE
HOSPITALIZATIONS OR DEATH
VIR BIOTECH
GILEAD
MERCK/RIDGEBACK
PFIZER
HOSPITALIZED SETTING
MORTALITY
MEDIAN TIME TO RECOVERY (DAYS)
CONVALESCENT PLASMA
DEXAMETHASONE
ROCHE
increase in symptomatic infections
increased hospital visits
increased deaths
increased mortality
increased recovery time

DRUGS THAT TREAT INFECTED PATIENTS

Some drugs target the virus itself to make it easier for the immune system to clear the infection. Other drugs might protect organs from damage while the immune system fights the infection. Finally, some other drugs stimulate the immune response to help it beat the virus or dampen the immune response to prevent it from overreacting to the infection and causing the patient more harm than good.

In addition to treating a known infection, drugs can be used prophylactically (before confirmation of infection) and could make sense for people at high risk of infection (e.g., healthcare workers) to prevent infection; they can also be used to treat someone with a known SARS-CoV-2 infection even before they show signs of disease to try to keep the symptoms as mild as possible.

Viable prophylactic therapies must be safe, well-tolerated, and convenient to administer; if drugs are dangerous, hard to tolerate, difficult to administer (e.g., IV), or difficult to manufacture at scale, then they may be reserved for sicker patients and used in the hospital (e.g., once someone is admitted and put on oxygen).

If drugs can both increase survival and facilitate quicker discharge from the hospital then such drugs might mitigate our fears of infection sufficiently to lift social distancing requirements, since we would then know that hospitals can quickly help the people who need help to recover safely. However, only remdesivir has shown clear activity to date and received Emergency Use Authorization (not same as full FDA approval, but for now may as well be the same), but it is hardly a cure (~30% reduction in time to discharge and likely a reduction in chance of death) and the fact that it requires IV administration and has potentially serious liver toxicity will limit its use to the hospital setting. Other drugs that seemed promising early on have since disappointed when we got more clinical data.

In terms of drugs that could help treat COVID-19 quickly and easily to allow society to open up again, the most promising classes of drugs for which we will have clinical data in 2020 are monoclonal antibodies (temporarily mimic what vaccines do) and oral polymerase inhibitors, (hopefully do what remdesivir does but better and more conveniently).

SARS-COV-2 REPLICATION AND TRANSMISSION

The virus that causes COVID-19 is called SARS-CoV-2. SARS stands for "severe acute respiratory syndrome," which can involve shortness of breath, pneumonia and/or respiratory distress, and the virus is so named because it closely resembles the virus (SARS-CoV) that caused the SARS outbreak of 2002/2003. Both viruses come from the coronavirus (CoV) family.

SARS-CoV-2 is enclosed by a protective lipid (fatty) membrane containing proteins that give the virus structure and allow for infection. The membrane is a weak point that makes it easy to kill with soap, bleach, or alcohol. Inside the virus there is a single strand of ribonucleic acid, or RNA, that contains the virus's genetic information. The coat of the virus consists of several structural proteins, including the spike protein (S), which allows the virus to attach to host cells and gives the virus its distinctive shape.

SARS-CoV-2 enters host cells by binding a specific protein, called "ACE2," on the cell surface; once inside, the virus hijacks the host's cellular machinery to replicate and assemble more viral particles. After the virus binds the cell, enzymes produced by the host (TMPRSS2) activate the viral spike protein (S), which allows the virus to enter the cell (see schematic below)[1,2]. Once inside the cell, the virus rapidly makes more copies of its genetic material (RNA) as well as the proteins needed to create new viral particles. The RNA is packed into new particles, which are released into the body to infect surrounding cells.

SARS-CoV-2 is thought to first infect the cells of the nose and throat, from where it can spread, but it does not cause significant symptoms until it progresses into the lung. During the first stage of infection patients are more likely to spread the virus to others by coughing or breathing, even before they start to show symptoms (referred to as "asymptomatic") or while their symptoms are too mild to distinguish from a harmless cold. Once the virus makes it way into the lungs, more notable symptoms (coughing, fever, and/or shortness of breath) start to become more prevalent. SARS-CoV-2 also uses additional mechanisms to slow down the response of the immune system, meaning that the infection can linger for longer and the host can infect more people. In contrast, SARS-CoV and MERS-CoV primarily and quickly infect lung cells, which are deeper in the respiratory tract and cause an earlier onset of serious symptoms, so patients can be identified and quarantined sooner before they spread the virus, making it easier to stamp out the original SARS and MERS cases before they triggered a pandemic.

1. Spike protein on the virion binds to ACE2, a cell-surface protein. TMPRSS2, an enzyme, helps the virion enter
2. The virion releases its RNA
3. Some RNA is translated into proteins by the cell's machinery
4. Some of these proteins form a replication complex with the viral RNA genome to make more copies of the genome, a process that introduces some mutations. Although the virus has proof-reading proteins that try to correct the mistakes, some slip through
5. Proteins and RNA are assembled into a new virion and
6. released

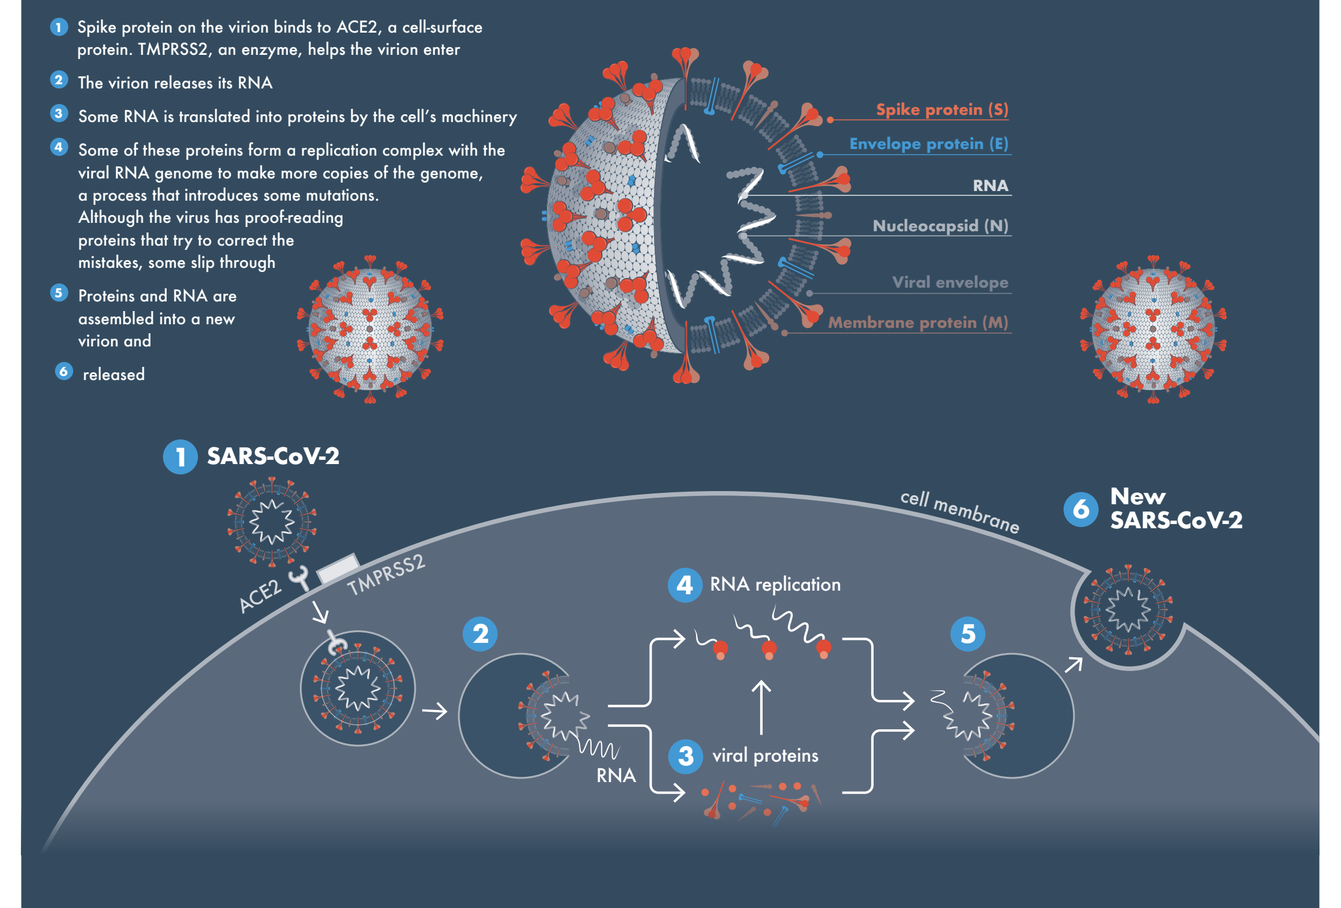

Spike protein (S)
Envelope protein (E)
RNA
Nucleocapsid (N)
Viral envelope
Membrane protein (M)
1 SARS-CoV-2
ACE2
TMPRSS2
cell membrane
2
3 viral proteins
RNA
4 RNA replication
5
6 New SARS-CoV-2

TRANSMISSION:

SARS-CoV-2 is spread from person to person mainly through respiratory droplets that are created when a person coughs, sneezes, or talks (similar to how influenza spreads). These droplets are thought to travel no more than six feet, thus this distance is reflected in current social distancing rules, though research shows microdroplets can travel further and therefore 6 feet should be considered a minimum. 10 feet apart is better. Wearing masks in public places can help prevent both the spread of these droplets to others if you are infected and at least partially reduce inhalation of these droplets from others if you are well.

SARS-CoV-2 may also spread by touching an infected surface followed by subsequent contact with the eyes, nose, or mouth. Regular hand washing and disinfection of commonly touched surfaces can help reduce this type of spread.

There is no evidence that SARS-CoV-2 can spread through food (different from foodborne viruses like norovirus or hepatitis A) or through mosquitoes (different from mosquito-borne viruses like Zika or West Nile). SARS-CoV-2 causes respiratory symptoms that increase its spread to others, unlike foodborne viruses that cause gastrointestinal symptoms and spread primarily through contact or sharing food or utensils. Furthermore, in contrast to mosquito-borne viruses, the spread of SARS-CoV-2 occurs directly from person to person and not through an animal host.

SARS-CoV-2 could theoretically be transmitted by blood transfusions, but viral transmission has not been linked to blood transfusion for SARS-CoV-2, SARS, or MERS. Viral genomic material has been detected in the blood of ~15% of patients hospitalized with COVID-193, but not in patients with milder disease[4]. For COVID-19, we do not yet know whether these patients have infectious viral particles in their blood or only (noninfectious) genomic material. Both SARS and MERS, two other related respiratory diseases caused by novel coronaviruses, were also associated with detectable virus particles in the blood during infections, particularly SARS (~75% of patients with SARS[5] and ~33% of patients with MERS[6]). Additionally, replicating coronavirus particles have been detected in the blood of mice infected with SARS[7]. We also know that non-human primates can be infected with the virus that causes SARS (SARS-CoV) via blood transfusions and that these animals also develop SARS symptoms[8]. While it is encouraging that none of these diseases have been linked to human blood transfusions, there are reasons to remain vigilant: SARS and MERS had very different infection numbers (~8,000 SARS infections[9] and ~2,500 MERS infections[10] worldwide) and almost no asymptomatic infections (greatly reducing the risk of infected patients donating blood).

DISCLAIMER

RA Capital Management, LP ("RA Capital") is an investment adviser registered with the Securities and Exchange Commission ("SEC") under the Investment Advisers Act of 1940, as amended ("Advisers Act").

From time to time, RA Capital may hold long and/or short positions in companies which are included in this map. The information contained in this map should not be interpreted or construed as investment or other professional advice. All information about the individual companies referenced in this map is presented for the purpose of progressing a strategic approach to the Coronavirus Disease 2019 ("COVID") crisis, and is no way inclusive of all the factors that should be considered when making an investment decision. Such considerations include, but are not limited to, the financial condition of a firm, the skill of the management team, the evolving competitive landscape, and other revenue sources outside of COVID-related projects. Therefore, nothing herein should be used to partially or fully inform an investment decision. The map is provided for informational purposes only, and nothing contained herein constitutes a solicitation, recommendation, endorsement, offer or an inducement to invest with RA Capital or its funds, or to buy or sell any securities or other financial instruments in any jurisdiction. The factual information contained within this map is publicly available. The analysis of such data represents the views held by RA Capital and its employees at the time of publication and is subject to change without notification. Nothing in this map should be considered medical advice or guidance on the treatment or prevention of COVID.

Target the Virus Itself

Target viral replication
By preventing the virus from infecting host cells or replicating once inside the cell, these drugs could prevent patients from developing severe disease and help resolve the infection sooner.

We understand how these drugs work, making it easier to predict their success or failure in trials and to extrapolate findings to other similar drugs
This is a big advantage for predicting clinical trial results; when you do not know how a drug works, doing a clinical trial is more of a shot in the dark.

Prevent viral entry into cells, possibly preventing infection (if given before someone has been exposed) or reducing disease severity by preventing the virus from spreading from cell to cell once a person is infected
The antibodies in this section also help target infected cells for destruction by the immune system.

More likely to work: drugs specific for blocking SARS-CoV-2 entry
Viral entry requires several steps:
1. The viral Spike protein must bind to a receptor called ACE2 on the cell surface in the right way (to its receptor binding domain, or RBD). Drugs that lower ACE2 or block the ACE2/Spike interaction might therefore prevent the virus from attaching to cells.
2. After binding ACE2, an enzyme called TMPRSS2 on a cell's surface must 'clip' the viral Spike protein, activating it. Drugs that inhibit TMPRSS2 might therefore interfere with this process.
3. The virus fuses with the host cell membrane and enters the cell.

Target the virus to block viral entry

Prevent virus from attaching to cells while also targeting infected cells for immune destruction using antibody therapies
- Antibodies can both bind to the virus to prevent it from attaching to cells and also direct the immune system to destroy infected cells (which have viral proteins on their surface).
- Monoclonal antibodies can be engineered to only block the virus from infecting cells without targeting infected cells for destruction, but it is not clear whether any company has chosen to disable this "effector" function. If they have, that would potentially make the monoclonal antibody less effective.
- As more information becomes available on the design of various antibody therapies we can classify them according to whether they only block viral entry or whether they both block entry and have immune effector functions. Even among antibodies that have immune effector functions, there are those that have ordinary levels of effector functions and those that have been engineered with increased effector function (e.g., Vir made both versions of its lead antibody).
- Polyclonal antibodies (poly means 'many,' as opposed to mono, meaning 'one') are mixtures of antibodies derived from a person or immunized animals. One strategy is using convalescent serum, which means taking the (polyclonal) antibodies from a patient who has recovered from COVID-19 (i.e., has convalesced) and giving them to help other patients clear the infection. These antibodies are not typically modified from their natural form, though it is technically possible to engineer them further to increase potency. Currently no companies we are aware of are modifying polyclonal antibodies for treating COVID-19. Another strategy is to immunize animals, (e.g., horses or cows), to use their antibodies; however, they will first require developing a vaccine of some kind that can be given to animals.

Monoclonal antibodies against Spike protein that could both neutralize the virus and direct the immune system to destroy infected cells: likely to work (since they mimic how the immune system fights the virus) but will take time to produce at very large scale (for millions and tens of millions of patients)
- There is good reason to believe that this drug class could work and might be clinically validated. FDA approved, and ready for at least broad-scale use by end of 2020, maybe even late Fall 2020.
- Antibodies will be tested for treating infections and as prophylaxis to prevent infection in those at risk. If used to prevent infection, they would likely be limited to front-line workers due to limited supply; alternatively, they could be provided to vulnerable members of households, or nursing homes, where increased infection risk is suspected (e.g., vulnerable individuals who live with essential workers). For prophylaxis, IV delivery of the antibody would be too inconvenient for large numbers of people. Therefore, if we aim to provide prophylaxis to a larger proportion of the community, the ideal prophylactic antibody product would need to be injected (up to 2 ml, can fit in a subcutaneous injection, up to 3 mL can fit into an intramuscular one; antibodies can be formulated to a concentration of 100-150 mg/mL, so in the worst case a person could get more than one injection at a time).
- Some companies have evaluated antibodies produced by the immune systems of recovered patients to identify specific antibodies that appear most effective at (1) neutralizing the virus by binding to viral Spike protein and thus blocking viral entry into cells and (2) directing the immune system to destroy infected cells.
- These antibodies need to be produced at large scale but manufacturing will not meet global demand if these agents are effective. Estimating that each patient is likely to need around 0.3-1.5 grams, treating millions of people would require producing literally tons of antibody material, meaning that even if these agents work we will still need to flatten the curve (through social distancing and other basic protective measures) to make the most of what we can produce.
- While it is possible that giving a dose to an uninfected person could protect them from infection for many months (~6 months is plausible), they are no substitute for a vaccine. One metric ton would make 1M doses of 1 gram each, and the most widely used antibody products are manufactured on the scale of tens of metric tons per year, not hundreds.

Combination of different neutralizing antibodies — **Advancing an antibody cocktail: more likely to withstand emergence of mutations**

- *EUA* Regeneron/Roche (REGEN-COV/(casirivimab + imdevimab)) IV/SUBQ BARDA — *EUA revised due to omicron variant | High risk, outpatient setting and post-exposure prophylaxis (PEP) setting
- *EUA* Eli Lilly/AbCellera Biologics/Junshi Biosciences (LY-CoV555/bamlanivimab + LY-CoV016/etesevimab) IV — *EUA revised due to omicron variant | High risk, outpatient setting and post-exposure prophylaxis (PEP) setting
- EUA AstraZeneca (Evusheld/tixagevimab+cilgavimab) IM
- POSITIVE TOP-LINE DATA Brii Biosciences (BRII-196+BRII-198) IV
- National Resilience/Ology Bioservices (ADM03820) SUBQ
- BMS/Rockefeller University (BMS-986414 + BMS-986413) IV/SUBQ
- Sorrento Therapeutics (COVISHIELD)
- Aridis (AR-701) IV
- Aridis (AR-712/AR-711+AR-720) INHALED

Single multi-valent molecule that binds different sites on Spike
Only current example lacks effector functions, which may reduce efficacy relative to some other antibody therapies
- POSITIVE TOP-LINE DATA Novartis/Molecular Partners (MP0420/ensovibep) IV
- Novartis/Molecular Partners (MP0423)

Broadly neutralizing antibodies: may be effective against other coronaviruses and more resistant to SARS-CoV-2 variants
- *EUA* Vir Biotechnology/GSK (VIR-7831/sotrovimab) IV/IM — *Outpatient setting
- Adagio Therapeutics (ADG20) IM
- Eli Lilly/AbCellera Biologics (bebtelovimab/LY-CoV1404) IV — *Testing as a monotherapy and in combo with bamlanivimab and etesevimab
- Exevir (XVR-011) IV
- Vir Biotechnology/GSK (VIR-7832*) IV

Advancing single antibodies binding one site on the Spike protein: increased risk of mutations that could reduce antibody binding
Several of these individual antibodies could be combined if needed.

Specific for SARS-CoV-2: may not be effective against other coronaviruses and potentially vulnerable to SARS-CoV-2 variants
- *EUA REVOKED* Eli Lilly/AbCellera Biologics (LY-CoV555/bamlanivimab) IV — *As monotherapy, due to loss of potency against emerging variants
- POSITIVE DATA* Celltrion (CT-P59/regdanvimab) IV — *outpatient setting, conditionally approved in South Korea & Europe
- Tychan (TY027) IV
- SinoCelltech (SCTA01) IV
- Sorrento Therapeutics (COVI-AMG/STI-2020) IV
- Abpro/Mabwell (ABP 300/MW33) IV
- AbbVie/Harbour BioMed (ABBV-47D11) IV
- AbbVie (ABBV-2B04) IV
- CORAT Therapeutics/Yumab (COR-101) IV
- Jemincare (JMB2002) IV
- Hengenix/Sanyou/Shanghai ZJ Bio-Tech (HLX70) IV
- Luye Pharma (LY-CovMab) IV
- Sorrento Therapeutics (COVI-DROPS/STI-2099) INTRANASAL
- BeiGene/Singlomics Biopharmaceuticals (BGB-DXP593) IV
- IGM Biosciences (IGM-6268) INTRANASAL
- Adaptive Biotechnologies
- ImmunoPrecise Antibodies (TATX-03b)
- IDBiologics/Vanderbilt University Medical Center
- Eureka (Invisimask) INTRANASAL
- HiFiBio/ABL Bio (HFB30132A) IV
- Alphamab/Institut Pasteur Shanghai
- Sorrento Therapeutics (COVI-GUARD/STI-1499)
- Boehringer Ingelheim/University of Cologne (BI 767551/DZIF-10c) INHALED

Polyclonal antibodies derived from animals: not currently available but have potential for greatly increased scalability
Can produce tens of thousands of doses.

Engineered animals — Animals that produce human antibodies can avoid risk of developing immune response against the horse antibodies.
- **Engineered cattle**: SAB Biotherapeutics/CSL Behring (SAB-185) BARDA IV — *Positive Ph2 data - moving to Ph3
- Xenothera (XAV-19) IV

Non-engineered animals
- **Pigs**
- **Horses** — Risk of developing unwanted immune response against the horse antibodies.
 - Emergent BioSolutions (COVID-EIG) IV
 - Inmunova (INM005)* — *Approved in Argentina

Polyclonal antibodies: less scalable if derived from people than from animals
It is not yet clear whether antibodies taken from people who recovered from mild infections will be strong enough to protect people from serious infections.

Polyclonal antibodies purified from plasma of recovered patients: available now in small amounts but cannot be produced at large scale
- Convalescent serum contains a mixture of different (polyclonal) antibodies that could provide protection for people that have not yet generated their own immune response.
- The advantage of using convalescent serum is that it is available right now from everyone who has recovered from COVID-19. But these antibodies will first require evaluation in clinical studies to demonstrate how effective they are. For example, some labs are reporting that there are low neutralizing antibodies in the serum of patients who recovered from mild infections.
- If these polyclonal antibodies from recovered patients demonstrate that they save lives, then many people will be asked to donate their blood. It will be important to understand the right therapeutic dose of these antibodies and also what factors impact quality; for example, it is not clear whether it would be better to use antibodies from someone who had a serious case and got to high antibody levels or whether it would be good enough to use antibodies from people who recovered from a mild or asymptomatic infection, which are easy to find.

Does not require plasma to produce — In fact, this is better characterized as a diverse cocktail of monoclonal antibodies. Conceptually very interesting but have not demonstrated production at large scale.
- Immunome (IMM-BCP-01) IV
- Grifols/Gigagen (GIGA-2050) IV

Hyperimmune globulin: purified antibodies that are derived from pooled convalescent plasma

Requires plasma source to produce — Production can be pooled across many donors to make a more consistent product
- Kamada/Kedrion IV
- GC Pharma (GC5131A*) IV — *Development in South Korea
- Grifols* SUBQ BARDA
- CoVIg-19 Plasma Alliance** IV
- Emergent BioSolutions (COVID-HIG) IV BARDA — **Alliance between CSL Behring, Takeda, Biotest, BPL, LFB, and Octapharma for plasma supply. *Hyperimmune globulin from CoVIg-19 Plasma Alliance, Emergent BioSolutions, and Grifols were tested together in hospitalized patients in an NIH-sponsored Ph3 trial and failed. Grifols is testing in the outpatient setting

Approved for Emergency Use
Convalescent plasma: no additional purification needed, just transfer from one person to another, but efficacy can vary from donor to donor
One plasma donation yields 2-3 doses of drug and potency of the antibodies in the plasma can vary from person to person.
- APPROVED (convalescent plasma*) IV — *Failed to show benefit in NIH-sponsored Ph3 RCT

Specifically block viral entry into cells using soluble ACE2 decoy protein to bind viral Spike protein (preventing it from binding real ACE2 on cells): likely limited to the hospital setting due to IV administration
In theory this should be an effective strategy though the manufacturing of proteins can be difficult, which will preclude a large supply of this type of drug in the near term.
- **Recombinant ACE2**: Apeiron (APN01) IV; Sytimmune (SI-F019) IV; Formycon (FYB207); Sorrento Therapeutics (STI-4398/COVIDTRAP); Neoleukin Therapeutics (NL-CVX1)

Impair viral entry by reducing the levels of ACE2 on cells, essentially making the cells untouchable to the virus: limited by potential side effects
If ACE2 is needed for important functions in the human body then reducing the levels on the surface of cells may cause side effects; however, these effects would likely be manageable since treatment would be needed only briefly.
- **Directly reduce ACE2 levels using RNAi** — Historically it has been difficult to deliver RNA to the lung
 - **Inhaled: ensure drug gets preferentially into lung**: Vir Biotechnology/Alnylam Pharmaceuticals INHALED
 - **Unknown route of delivery**: pHion Therapeutics (PTX-G1)
- **Indirectly reduce ACE2 levels using hormonal therapy**
 - **Androgen receptor antagonist**: FAILED Kintor (GT0918/proxalutamide) ORAL
 - **Estrogen**: Mithra Pharmaceuticals (estetrol) ORAL

Prevent virus from binding host receptor — Binding to host receptors like ACE2 is necessary for viral entry

Block virus entry through secondary cellular receptors: likely less relevant
There are reports that the SARS-CoV-2 virus can bind to receptors besides ACE2; however, the relevance of these additional binding mechanisms is currently unclear.
- **AXL**: BerGenBio (bemcentinib) ORAL
- **CD147**: Jiangsu Pacific Meinuoke Bio Pharmaceutical (meplazumab) IV

Target the host to prevent viral entry

Inhibit host protease — Proteolytic cleavage of Spike protein is necessary for viral entry

Target key cell protease TMPRSS2, which clips viral Spike protein to activate it and facilitate viral entry
- **Repurposed small molecules** — Faster development timelines and known safety and tissue distribution profile
 - Sagent Pharma/Hikma (nafamostat mesilate) ORAL
 - **Nafamostat**
 - **Oral**: COVISTAT/Ensysce (nafamostat) ORAL
 - **Potential for use outside of hospital: easier to administer formulations** — **Inhaled**: Pfizer/Daiichi Sankyo/University of Tokyo/RIKEN (nafamostat) INHALED; Chong Kun Dang Pharmaceutical (CKD-314/nafamostat mesilate) IV
 - **Likely limited to hospital setting: original IV formulation**: Sun Pharma (nafamostat mesylate) IV
- **Novel agent/modality developed specifically for SARS-CoV-2** — Potential for better potency/selectivity but slower development timeline and increased risks due to novel technology
 - **Inhaled 'RNA interference' (RNAi) drugs: less likely to get into lung** — Historically, it has been very difficult to get oligonucleotides (what RNAi drugs are made of) into the lung. Vir Biotechnology/Alnylam Pharmaceuticals

Target cathepsin-L, a cysteine protease that may help facilitate viral entry: Selva Therapeutics (SLV213) ORAL

Target various serine proteases that may help facilitate viral entry: RedHill Biopharma (upamostat) ORAL

Less likely to work: drugs repurposed to try to block SARS-CoV-2 entry
- **Non-specifically block viral entry into cells with recombinant surfactant-like protein**: Airway Therapeutics (AT-100) INTRATRACHEAL
- **Drugs developed to prevent influenza entry** — The process by which influenza enters cells is different from the SARS-CoV-2 entry process, making it unclear whether these drugs can stop SARS-CoV-2 from infecting cells.
 - **Block viral fusion with host cells**: FAILED Pharmstandard (Arbidol/umifenovir) ORAL
 - **Remove sialic acid from host lung cells to prevent viral adhesion** — The importance of sialic acid for SARS-CoV-2 entry is currently unclear. FAILED Ansun Biopharma (DAS181) INHALED

Block viral replication once the virus is inside the cell

Broad spectrum small molecule antivirals that can block various viral processes and are more easily delivered to the lung

Target replication of viral RNA genome

Most promising: target viral RNA polymerase (which makes copies of the viral genome) using nucleoside/tide analogs, essentially messing up the viral assembly line by inserting incorrect parts into the mix

Previously developed for treating other viruses and unlikely to be a perfect fit for SARS-CoV-2

More convenient delivery: Oral or inhaled
- If shown to be effective, these drugs could be used outside of hospitals or could even be sent to people's homes for self-treatment.
- Could be available by Fall 2020 and could be scaled up to help millions of patients by early 2021 if effective.
- The key to efficacy will likely be getting a high enough dose into the lungs.
- **Oral**
 - EUA Ridgeback Biotherapeutics/Merck (EIDD-2801/molnupiravir) ORAL
 - Atea/Roche (bemnifosbuvir/AT-527) ORAL
 - COMPLETE Gilead (Descovy/emtricitabine + tenofovir alafenamide fumarate) ORAL
 - Henan Zhenshi Biotechnology (azvudine) ORAL
 - Bukwang Pharmaceutical (clevudine) ORAL
- **Favipiravir**: FAILED FUJIFILM Toyama Chemical/Appili Therapeutics (Avigan/favipiravir) ORAL
- **Remdesivir**: COMPLETE Jubilant Pharma (remdesivir) ORAL; Gilead (GS-5245) ORAL
- **Inhaled formulation of approved drug remdesivir** — May allow for remdesivir to be used in milder patients. DISCONTINUED Gilead (remdesivir) INHALED

Approved in both the hospitalized and outpatient settings
Remdesivir: cuts hospital stay by ~30%, likely reduces risk of death, must be given IV, and has some risk of liver toxicity. 87% relative risk reduction for hospitalization or death in the outpatient setting. While the RoD of remdesivir is not viable in the outpatient setting (3 IV infusions over 3 days), this data provides PoC for polymerase inhibitors in the outpatient setting.
- APPROVED Gilead (Veklury/remdesivir) IV

IV (and thus outpatient use is logistically challenging): Biocryst (galidesivir) IV

Block production of the nucleosides: limiting the availability of the building blocks needed for viral replication
- **Block guanosine production** — IMPDH inhibitor: COMPLETE Bausch Health (Virazole/ribavirin) INHALED; FAILED BioSig Technologies/ViralClear Pharmaceuticals (merimepodib) ORAL
- **Block pyrimidine production** — DHODH inhibitor: Clear Creek Bio (brequinar) ORAL; PTC Therapeutics (PTC299/emvododstat) ORAL; Rhizen (RP7214) ORAL; Immunic (IMU-838) ORAL

Block viral protein function

Target viral protease — Protease is a viral enzyme that cuts viral raw protein materials into active pieces

Target Coronavirus main protease (Mpro)
- **Developed for human coronavirus**: EUA Pfizer (Paxlovid/nirmatrelvir+ritonavir) ORAL; Shionogi (S-217622) ORAL; Pardes Biosciences (PBI-0451) ORAL; Enanta Pharmaceuticals (EDP-235) ORAL; Sorrento Therapeutics (STI-1558) ORAL; DISCONTINUED Pfizer (PF-07304814) IV
- **Repurposed molecules**
 - **CETP inhibitor originally developed for heart disease** — Dalcetrapib was shown to inhibit the Mpro protease in vitro. FAILED DalCor Pharmaceuticals (dalcetrapib) ORAL
 - **Veterinary drug with activity in feline coronavirus** — Developed to inhibit coronavirus 3CL protease and was increased likelihood of inhibiting SARS-CoV-2. Anivive Lifesciences (GC376)

Repurposed HIV protease inhibitors: Ascletis (ASC09 + ritonavir) ORAL; FAILED J&J (Prezcobix/darunavir + cobicistat)* ORAL — *No in vitro activity according to J&J; FAILED (lopinavir + ritonavir) ORAL

Block viral protein translation or processing
- **Block transport of viral proteins from nucleus to cytoplasm using a toxic anticancer drug**
 - **XPO1 inhibitor**: Karyopharm (Xpovio/selinexor) ORAL
 - **MEK inhibitor**: Atriva Therapeutics (ATR-002) ORAL
- **Inhibit specific host proteases (calpains) that may be essential for viral replication**: FAILED Blade Therapeutics (BLD-2660) ORAL
- **Block translation of viral proteins using a toxic anticancer drug**: PharmaMar (Aplidin/plitidepsin) IV; DISCONTINUED eFFECTOR Therapeutics (eFT226/zotatifin) IV
- **Block intracellular transport along microtubules using a toxic anticancer drug**: POSITIVE TOP-LINE DATA Veru (VERU-111) ORAL

RNAi that can inhibit production of viral proteins by directing the cell's defenses to cut viral mRNA into pieces; will need to be inhaled to target the lung
Historically, it has been very difficult to get oligonucleotides (what RNAi drugs are made of) into lung cells.
- Vir Biotechnology/Alnylam Pharmaceuticals (VIR-2703/ALN-COV) INHALED
- Arrowhead Pharmaceuticals (ARO-COV) INHALED
- Sirnaomics (STP908) INHALED

Unclear mechanism of action: we do not really know how these drugs work; their use for COVID-19 is based on anecdotal evidence of antiviral activity and they have thus far not shown clear efficacy
- This is a big disadvantage that makes it less likely that they will actually work when we test them in patients (though not impossible; we still do not know how some effective drugs for a variety of diseases work).
- We already have enough data from trials evaluating chloroquine and hydroxychloroquine to say that these drugs either do not work, hardly work, or maybe even cause more harm than good (especially when combined with other ineffective drugs).

Generate NO in the nasal cavity — NO has antiviral properties in laboratory experiments
- GeneOne Life Sciences (GLS-1200) INTRANASAL
- SaNOtize Research and Development (NORS) INTRANASAL/GARGLE — *Seeking EUA in UK and Canada

Antimalarial drugs
- **Tafenoquine**: COMPLETE Sixty Degrees Pharma ORAL
- **FAILED — Chloroquine derivatives** — These established drugs have known cardiac side-effects (can cause sudden cardiac death). Clinical trials have thus far failed to show any efficacy for this class of drugs despite early signals from uncontrolled trials that suggested they helped patients recover faster.
 - **Better tolerated**: Genoscience Pharma (GNS561) ORAL
 - **Hydroxychloroquine**: FAILED (hydroxychloroquine) ORAL
 - **Less well tolerated** — Common side effects include stomach pain, nausea, vomiting and headache. FAILED (chloroquine) ORAL
 - **Administered together with an antibiotic to prevent bacterial pneumonia (but also as anti-inflammatory)**: FAILED (azithromycin)* ORAL — *+ chloroquine

Antihelminthic drugs
These drugs normally kill tapeworms, which are very unlike viruses, by poorly defined mechanisms, but there has been some in vitro evidence of efficacy against SARS-CoV-2, though not at doses that can be achieved in humans.
- **Niclosamide** — Commonly taken as an oral pill but not absorbed well into the bloodstream; this works well for killing gut-residing tapeworms but greatly reduces systemic drug levels in humans.
 - **Oral: unlikely to get much drug into lungs because of poor systemic absorption**
 - **Standard formulation**: NeuroBo Pharmaceuticals (ANA001/niclosamide) ORAL
 - **Gut targeted oral formulation**: First Wave BioPharma (niclosamide) ORAL
 - **Inhaled: more likely to get drug to the lungs though still lacks mechanistic rationale**: TFF Pharmaceutical (niclosamide) INHALED; COMPLETE Union Therapeutics (UNI91104/niclosamide) INHALED
 - **Intranasal**: Union Therapeutics (UNI91103/niclosamide) INTRANASAL
- **Broad spectrum antiparasitic agent** — These compounds are rapidly eliminated from the body and will likely need to be modified to achieve sufficiently high drug levels in the lung.
 - **Nitazoxanide** — **Extended release formulation to increase drug exposure**: FAILED Romark Laboratories (NT-300/nitazoxanide) ORAL — *Failed its PIII, but is seeking EUA
 - **Ivermectin** — **Extended release formulation to increase drug exposure**: MedinCell (ivermectin) ORAL; **Standard formulation**: FAILED (ivermectin) ORAL

Other repurposed drugs with unclear antiviral MOAs — Predicted antiviral activity from laboratory experiments.
- **Repurposed anti-depressant**: FAILED (fluvoxamine) ORAL — *Positive results in a small Ph2 RCT
- **Upregulate miR-124** — Reduces expression of pro-inflammatory cytokines and shown to reduce replication of SARS-CoV-2 in vitro. FAILED Abivax (ABX-464) ORAL
- **Repurposed antibiotic**: FAILED (azithromycin) ORAL

COVID-19 THERAPEUTICS: TARGET VIRAL REPLICATION + CELLULAR ENTRY MILESTONES* (DATA AS OF FEBRUARY 11, 2022)

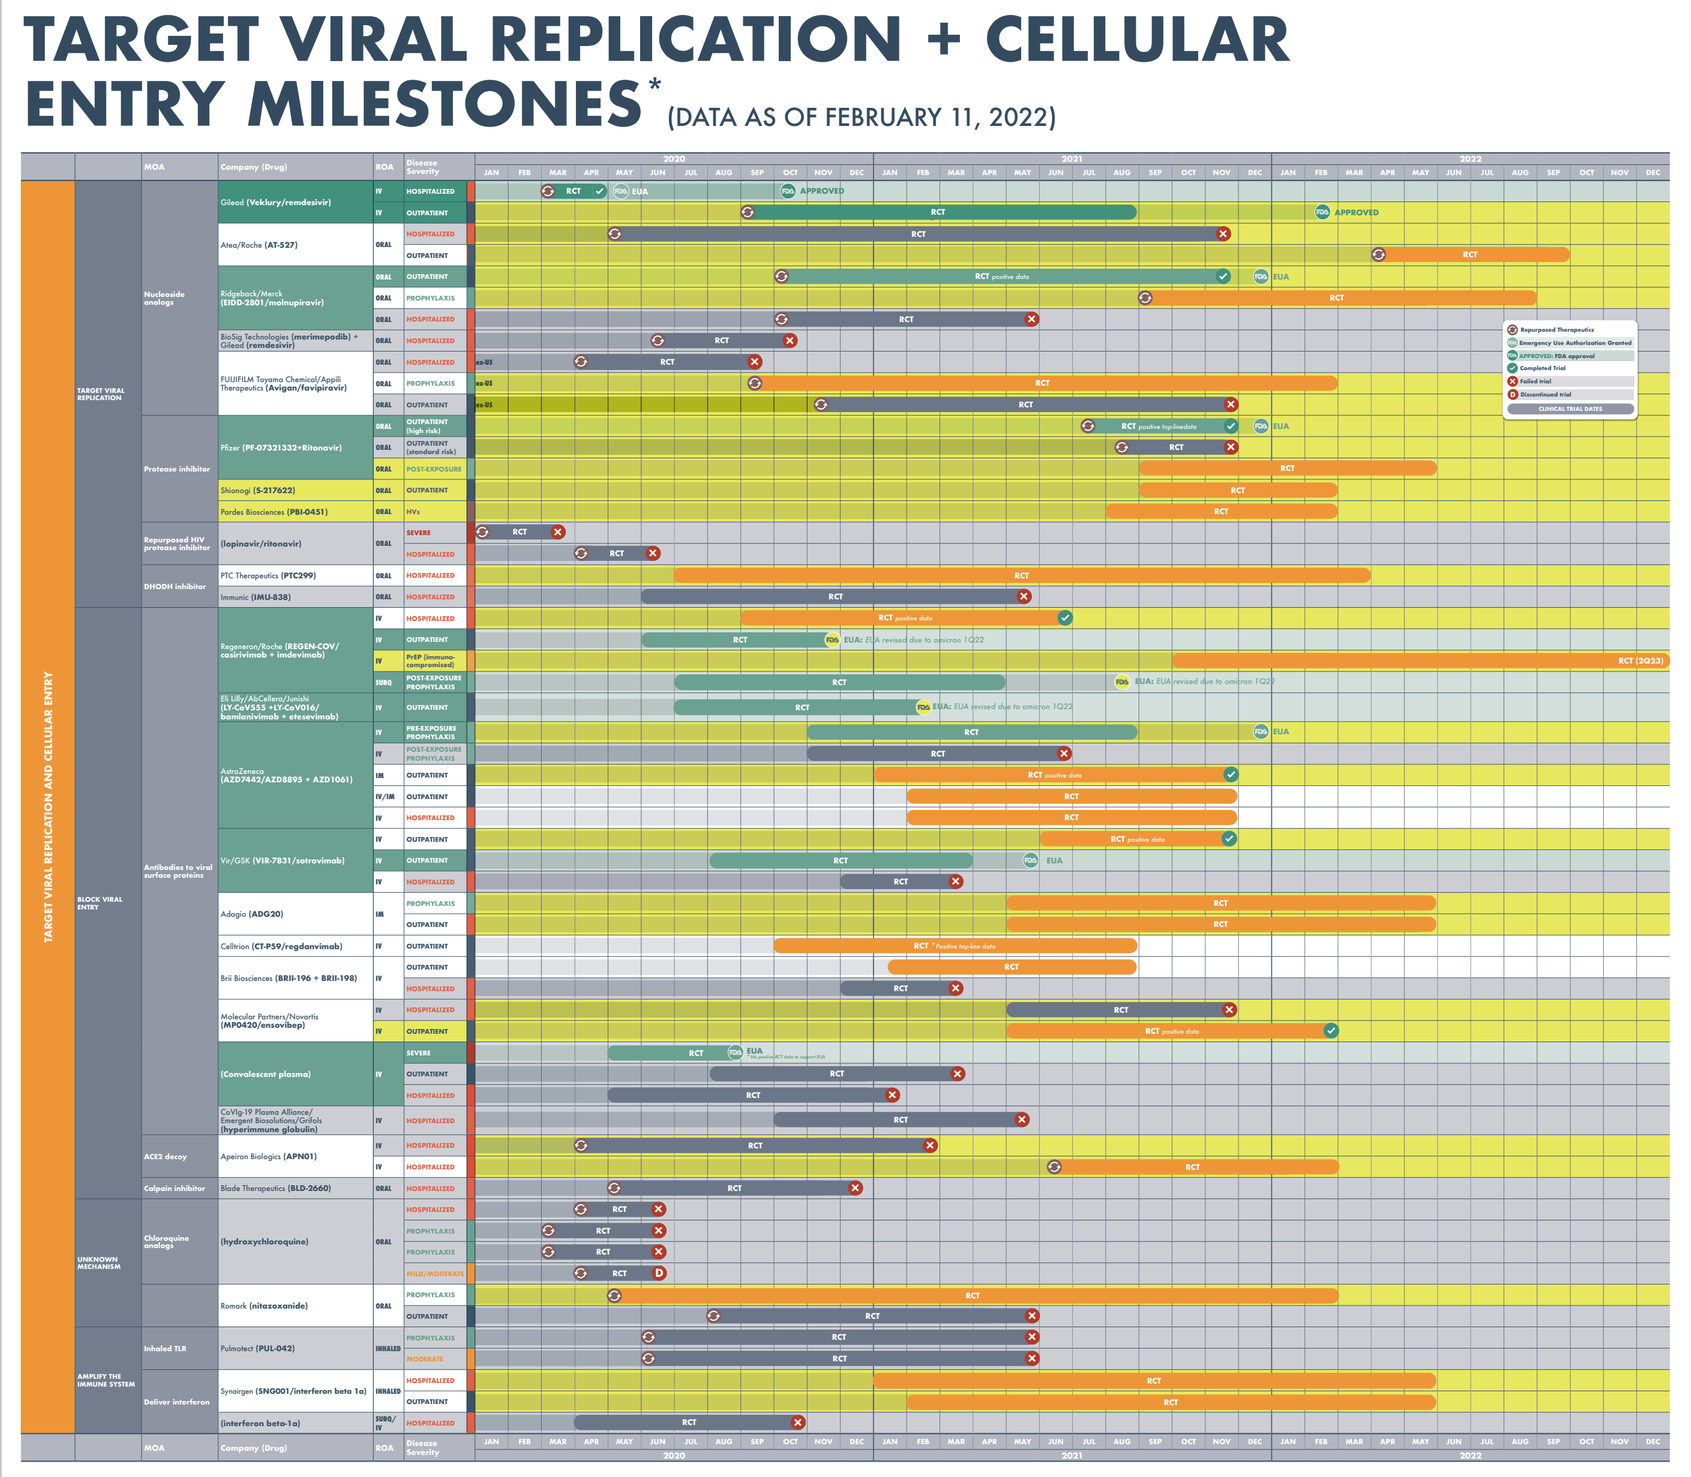

* Milestone dates are approximated based on the estimated primary completion date as reported on clinicaltrials.gov or our best estimate for when we could see data

Target the immune response against the virus to help clear the infection

Success of these programs in animal models may not predict success in humans, partly due to differences in immune responses across organisms. We might therefore discount the probability of success of many of these programs.

Stimulate immune response — Some of these approaches may be risky (inhaled immune stimulants), or tedious and hard to manufacture at scale (infused NK cells), and will therefore require evidence that they will not cause excessive inflammation resulting in lung damage.

Deliver conventional therapeutic drugs: easier to manufacture at scale

Deliver immune stimulatory factors
- **Interferon**
 - **Interferon alpha**
 - **Inhaled**: Altum Pharmaceutical/BetterLife Pharma (AP-003/Interferon α2b) INHALED
 - **Injected**: Zydus Cadila (Virafin/peginterferon alfa-2b) SUBQ — *Approved in India
 - **Interferon beta**
 - **Inhaled**: Synairgen (SNG001/Interferon beta 1a) INHALED
 - **Injected**: Faron (Traumakine/interferon beta-1a) IV; FAILED Merck KGaA (Rebif/interferon beta-1a) SUBQ
 - **Interferon lambda** — Rationale is to stimulate the immune response in mild patients in whom drug is less likely to exacerbate cytokine storm (though drug is long-acting). Fewer receptors for interferon-lambda in the lung than other interferons, so potentially safest of the interferons. Eiger BioPharmaceuticals (Peginterferon Lambda-1a) SUBQ
- **TLR agonists**
 - **Inhaled TLR agonists**
 - **TLR3**: AIM Immunotech (Ampligen/rintatolimod*) IV — *NDA rejected by FDA for chronic fatigue syndrome
 - **TLR 2/6/9**: COMPLETE Pulmotect (PUL-042) INHALED
 - **Intranasal TLR4 agonists**: DISCONTINUED Revelation (REVTx-99) INTRANASAL
- **Interleukin (IL)**
 - **IL-2 agonist**: COMPLETE Nektar (NKTR-214/bempegaldesleukin) IV
 - **Deliver IL-7**: Revimmune (CYT107) IM; Genexine/NeoImmuneTech (NT-I7/GX-I7) IM
- **GM-CSF**: Partner Therapeutics (Leukine/sargramostim) INHALED/IV

Inhibit suppressive factors
- **Anti-CD73 antibody** — Activates B cells and induces antibody production. FAILED Corvus (CPI-006) IV
- **TGF-β2 knock down** — TGF-β2 suppresses the immune system, thus its inhibition may have an immunostimulatory effect. COMPLETE Oncotelic (OT-101) IV
- **Inhibit mTORC** — Upregulate innate antiviral gene expression. FAILED Adicet Bio/resTORbio (RTB101) ORAL

Deliver unconventional therapeutic modality: harder to manufacture at scale
- **Deliver NK cells: limited to hospitals because of supply constraints** — Infuse NK cells to promote antibody-mediated killing of infected cells. MiNK Therapeutics/Agenus (AgenT-797) IV; DISCONTINUED Celularity/Sorrento Therapeutics (CYNK-001) IV
- **Deliver adenovirus vector**: DISCONTINUED Altimmune (T-COVID) INTRANASAL

Protect the lung from injury while natural immune system fights infection

Goal is to help patients weather the infection long enough for the immune system to clear it

Target the Renin-Angiotensin System (RAS)
- SARS-CoV-2 enters lung cells via the ACE2 receptor, a membrane-bound enzyme that lowers blood pressure by helping convert angiotensin II (AngII) into angiotensin-(1-7). Angiotensin-(1-7) is a peptide that acts through the MAS receptor to maintain an antiinflammatory state.
- When SARS-CoV-2 enters the cell, it takes the ACE2 receptor with it, lowering ACE2 levels and causing AngII build up. Blocking the pro-inflammatory AngII receptor AT1, or agonizing the antiinflammatory AT2 or MAS receptors may overcome this imbalance and reduce lung inflammation. Similarly, blocking ACE may lead to increased levels of angiotensin-(1-7), the product of ACE2-cleaved AngII.

Activate receptors that promote vasodilation and antiinflammatory state
- **AT2 receptor agonist**: Vicore Pharma (VP01/C21*) ORAL — *Planning Ph3
- **MAS receptor agonist**: Constant Therapeutics (TXA127/Angiotensin 1-7) IV; Biophytis (Sarconeos/BIO101) ORAL
- **Inhibit ACE** — ACE inhibitors; Increase levels of Angiotensin (1-7). (ramipril) ORAL

Resist overactivation of pro-inflammatory AT1 receptor
- **AT1 receptor antagonist** — Angiotensin II receptor blockers (ARBs). (losartan) ORAL; (telmisartan) ORAL; (valsartan) ORAL
- **AT1 receptor agonist** — Paradoxically, counteracts overactivation of AT1 receptor. Trevena (TRV027) IV

Deliver the cytokine IL-22 — IL-22 helps protect tissue integrity and promotes repair
- Evive Biotech (F-652) IV
- DISCONTINUED Roche (UTTR1147A/RG7880) IV

Keep Severely Ill Patients Alive While Other Strategies (Natural Immunity and/or Antiviral Therapy) Clear Infection

- Goal is to extend life in the hopes that the patient's immune system and/or antiviral therapies can beat the infection.
- Patients with advanced stages of COVID-19 will often develop acute respiratory distress syndrome (ARDS) or broad organ failure.

Treat or prevent acute respiratory distress syndrome (ARDS)
- ARDS involves widespread inflammation that leads to respiratory failure.
- Goal is to reduce inflammation and improve oxygen saturation in the lungs to prevent the need for mechanical ventilation or manage those who are already on it.

Prevent onset or reduce impact of cytokine storm
- Acute viral disease can trigger an overreaction of the immune system, commonly referred to as a "cytokine storm," which can be fatal.
- Drugs aiming to prevent the onset of cytokine storm or reduce its impact generally suppress parts of the immune system to limit its overactivation while still leaving a sufficient immune response against SARSCoV-2 to clear the infection.

Inhibit specific cytokines — By inactivating specific pathways involved in recruiting the immune system to the lungs, these drugs are more likely to disarm harmful immune responses while keeping beneficial virus-specific responses intact. The trouble is that we are not yet sure which cytokines are most important in COVID-19, so we will learn from trials.

Target IL-6 signaling
- IL-6 is a known mediator of cytokine storm, though early data in COVID-19 suggests this strategy will not work well. Antibodies targeting IL-6 or IL-6R prevent soluble IL-6 from binding the IL6 receptor, reducing signaling and risk of cytokine storm. Antibodies like Actemra are already approved to treat the overactive cytokines associated with cancer therapies.
- Anti-IL6R mAbs such as Actemra and Kevzara have failed in multiple trials in COVID-19. However, a recent trial evaluating Actemra or Kevzara on top of dexamethasone in critically ill patients with COVID-19 showed reduced mortality (22%-28% vs. 36% with control) and increased freedom from organ support (median 10-11 days vs. 0 days with control), leading to approval in the UK.
- **Inhaled**: Tiziana Life Sciences (TZLS-501) INHALED
- **Subcutaneous**: Biocad (BCD-089/levilimab) SUBQ; R-Pharm/Cromos Pharma (olokizumab) SUBQ
- **IV**: Roche (Actemra/tocilizumab) IV; CSL Behring (clazakizumab) IV; J&J (Plivensia/sirukumab) IV; EUSA Pharma (Sylvant/siltuximab) IV; Sanofi/Regeneron (Kevzara/sarilumab) IV

Target GM-CSF signaling — GM-CSF is a cytokine associated with hyperinflamed organs, including the lung. Antibodies targeting GM-CSF can reduce GM-CSF signaling, which may help reduce inflammation.
- **Anti-GM-CSF antibody**
 - **lenzilumab** — In a RCT in hospitalized patients with COVID-19 (n=520), lenzilumab increased ventilator-free survival at 28 days on top of SoC (failure to achieve survival without ventilation: 15.6% lenzilumab vs. 22% pbo, p=0.0365, HR=1.54). However, lenzilumab did not impact the mortality endpoint in a stat sig manner, but showed only a positive trend favoring lenzilumab (9.6% lenzilumab vs. 13.9% pbo, p=0.2287). Humanigen (lenzilumab) IV — *Initial EUA declined. Awaiting additional data
 - I-MAB (TJM2/TJ003234) IV; GSK (otilimab) IV; Izana Bioscience (namilumab); Roivant Sciences (gimsilumab) IV; Nobelpharma (sargramostim) INHALED
- **Anti-GM-CSFRα antibody**: Kiniksa Pharmaceuticals (mavrilimumab) IV

Target interleukin-1 (IL-1) signaling — IL-1β is a pro-inflammatory cytokine implicated in driving lung inflammation and the onset of ARDS. Blocking IL-1 signaling may help reduce inflammation and associated lung injury.
- **IL-1 receptor antagonist protein**: Sobi (Kineret/anakinra) IV
- **Inhibit IL-1β**
 - **Anti-IL-1β antibody**: Novartis (Ilaris/canakinumab) IV
 - **IL-1β inhibitor fusion protein**: R-Pharm/Cromos Pharma (RPH-104) SUBQ

Target interferon gamma (IFNγ) — IFNγ is a proinflammatory cytokine believed to contribute to immune-mediated lung injury. Blocking IFNγ antibodies may help reduce inflammation. Sobi (Gamifant/emapalumab) IV

Target IL-8 signaling — IL-8 released by intra-alveolar macrophages enhances the recruitment of neutrophils and monocytes into the lung during ARDS. Blocking IL-8 signaling may help reduce hyperinflammation in the lung.
- **Anti-IL-8 antibody**: Bristol-Myers Squibb (BMS-986253) IV

Target IL-33 signaling — IL-33 is involved in regulating lung immune response in asthma. Blocking the binding of IL-33 to the ST2 receptor inhibits IL-33 signaling.
- **Anti-IL-33 mAb**: Roche (RG-6149/AMG-282) IV

Target LIGHT signaling — LIGHT is a novel cytokine implicated in activating both T and B cells and upregulates other inflammatory cytokines.
- **Anti-LIGHT antibody**: Avalo (AVTX-002) SUBQ

Target TNF-α signaling — TNF-α is a multifunctional cytokine implicated in hypoxia and onset of ARDS. Blocking TNF may help reduce inflammation.
- **Anti-TNF antibody**: (infliximab) IV
- **Inhibit soluble TNF**: Xencor/Inmune Bio (Quellor/XPro1595) SUBQ

Target CSF-1R signaling — CSF-1R signaling is associated with macrophage expansion and infiltration.
- **Anti-CSF-1R mAb**: Syndax (axatilimab/SNDX-6352) IV

Target IL-23 signaling — IL-23 induces the proliferation of Th17 cells. Blocking IL-23 signaling may help reduce inflammation. AbbVie (Skyrizi/risankizumab) IV

Corticosteroids
- This class of drugs works as anti-inflammatory agents, blocking the production of cytokines that drive the disease.
- Dexamethasone was shown to reduce mortality by 35% in ventilated patients and 20% in those requiring oxygen, leading to its approval in the UK and likely other countries in the near future. The success of dexamethasone raises the likelihood that other corticosteroids will also prove beneficial.
- **Dexamethasone**
 - **Standard dose**: (dexamethasone) IV
 - **High dose dexamethasone** — May induce NKT cell activation. AVM (AVM0703/dexamethasone) IV
 - ORAL/IV (methylprednisolone)
- **Additional antiviral activity reported**: Covis Pharma (Alvesco/ciclesonide) INHALED

Alkaloids — In a large RCT in non-hospitalized patients with PCR-confirmed COVID-19 (n=4159), the alkaloid colchicine reduced the composite rate of death or hospitalization (PEP) compared to placebo (4.6% vs. 6%; p=0.04). In the ITT analysis (n=4488), which included patients who were not PCR-confirmed, the benefit with colchicine was not stat sig for the PEP, but showed a positive trend on this outcome relative to placebo (4.7% vs. 5.8%; p=0.08). ORAL (colchicine)

Agents that are broadly immunosuppressive
- **JAK inhibitors**
 - **Inhaled** — Allows for more drug in the lung and less throughout the body, hopefully avoiding side-effects of this class. Theravance Biopharma (TD0903) INHALED
 - **Oral**
 - **pan-JAK inhibitor**: Pfizer (Xeljanz/tofacitinib) ORAL
 - **Approved for Emergency Use** — **Additional purported antiviral effect by blocking viral entry**
 - Baricitinib in combination with remdesivir was shown to reduce mortality by 43% relative to remdesivir in patients requiring high-flow oxygen/non-invasive ventilation and by 60% in those requiring oxygen.
 - Further, baricitinib in combination with remdesivir was shown to reduce median recovery time by 1 day compared to remdesivir in hospitalized COVID-19 patients (7 days vs 8 days).
 - Eli Lilly (Olumiant/baricitinib) ORAL EUA
 - **JAK2/FLT3 inhibitor** — Also inhibits IRAK1 and CSF1R. CTI Biopharma (Enpaxiq/pacritinib) ORAL
 - Incyte/Novartis (Jakafi/ruxolitinib) ORAL
- **Opioid receptor antagonist**: Cytocom/Immune Therapeutics (Lodonal/low dose naltrexone) IV

Agents that reduce immune cell trafficking
- **Anti-CCR5 antibody**: CytoDyn (leronlimab) SUBQ
- **Neutrophil adhesion receptor blockers**: Arch Biopartners (Metablok) IV
- **N-methyl-D-aspartate (NMDA) receptor antagonist**: Algernon Pharma (ifenprodil)
- **CCR2 antagonist**: Dimerix (DMX-200) ORAL
- **Anti-CXCL10 antibody**: AbbVie (Cenicriviroc) ORAL
- **CCR2 and CCR5 inhibitor**: Edesa Biotech (EB06) IV

Agents targeting the complement system
- **Inhibit C5/C5a/C5aR**
 - **Anti-C5 antibody**: Alexion (Ultomiris/ravulizumab) IV
 - **Anti-C5a antibody**: InflaRx (IFX-1) IV
 - **Anti-C5aR antibody**: Innate Pharma (IPH5401/avdoralimab) IV
 - **Peptide inhibitor of C5**: UCB (zilucoplan) SUBQ
- **Inhibitory C1-INH protein**: Pharming (Ruconest/recombinant C1 esterase inhibitor) IV
- **Inhibitory C1q peptide**: ReAlta Life Sciences (RLS-0071) IV
- **Inhibit C3**: Amyndas (AMY-101) IV
 - **Inhibitory C3 peptide**: Apellis (APL-9) IV
 - **Inhibitory C3 antibody**: NGM Biopharma (NGM-621) IV
- **Inhibitory C2 antibody**: Argenx (ARGX-117) IV
- **Inhibitory MASP-2 antibody**: Omeros (narsoplimab) IV

Target other aspects of inflammation
This is a catch-all category for a wide range of drugs, some of which broadly inhibit inflammation (and therefore might have more side-effects) and others which work on a specific branch of the immune system. It is hard to say which is more likely to work, hence the need for many studies. As trials read out, we can fill in this section to see if patterns emerge that might help us understand the nature of the im

Immuno-suppressive cell therapies
- **Stem cell approaches**: Athersys/Healios (MultiStem/HLCM051) IV; Celltex Therapeutics (AdMSCs) IV; Orbsen Therapeutics (ORBCEL) IV; Capricor (CAP-1002) IV; Stemedica (Stemedyne-MSC) IV; Sorrento (COVI-MSC) IV; Cartesian Therapeutics (Descartes-30) IV; Aspire (ACT-20) IV; Celodrus (CLBS119) IV; NantKwest (BM-Allo.MSC) IV; Novartis/Mesoblast (remestemcel-L) IV; Pluristem (PLX-PAD) IM
- **Protective probiotic bacteria**: Evelo (EDP1815) ORAL; Kaleido Biosciences (KB109) ORAL; 4D Pharma (MRx-4DP0004) ORAL
- **Treg cells**: Cellenkos (CK0802) IV
- **Apoptotic cells** — Dampen macrophage/dendritic cell cytokine signaling. Enlivex Therapeutics (Allocetra) IV

Other immunomodulatory approaches

More convenient delivery — Oral or intranasal
- **BTK inhibitor** — Reduces cytokine production broadly. AbbVie/J&J (Imbruvica/ibrutinib) ORAL; AstraZeneca (Calquence/acalabrutinib) ORAL; BeiGene (Brukinsa/zanubrutinib) ORAL
- **Tyrosine kinase inhibitors**
 - **Inhibit c-Kit**: AB Science (masitinib)+ isoquercetin ORAL
 - **Inhibit Bcr-Abl**: Exvastat (imatinib) ORAL
 - **Inhibit EGFR and BTK**: Sorrento Therapeutics (STI-5656/abivertinib maleate) ORAL
 - **Inhibit Syk**: Rigel (Tavalisse/fostamatinib) ORAL
- **Calcitonin gene-related peptide (CGRP) receptor antagonist** — Drug developed for migraine that may block upregulation of IL-6 and other inflammatory cytokines. Biohaven (BHV-3500/zavegepant) INTRANASAL
- **Neurokinin-1 antagonist** — Reduce neurogenic inflammation triggered by release of neuropeptides. Vanda Pharmaceuticals (tradipitant) ORAL
- **MIF, PDE4, and PDE10 inhibitor** — Asthma drug with anti-inflammatory properties. MediciNova (ibudilast/MN-166) ORAL
- **RASP inhibitor** — Reduce production of inflammatory cytokines. Aldeyra (ADX-629) ORAL
- **LSD1 inhibitor** — Decrease levels of inflammatory cytokines. Oryzon (vafidemstat) ORAL
- **PDE4 inhibitor** — Psoriasis and psoriatic arthritis drug with anti-inflammatory properties. Amgen (Otezla/apremilast) ORAL
- **PDE3/PDE4 Inhibitor** — Repurposed COPD drug with anti-inflammatory and bronchodilator activity. Verona Pharma (ensifentrine) INHALED
- **Adenosine A3 receptor agonist** — Inhibits TNF-α production via adenosine. Can-Fite (CF101/piclidenoson) ORAL
- **α1-adrenergic receptor antagonist** — Repurposed blood pressure drug, which could inhibit production of IL-6 and other cytokines via α1-adrenergic receptor signaling. (prazosin) ORAL
- **Sphingosine kinase-2 (Sk2) inhibitor** — Inhibits the formation of S1P, thereby reduced TNF-α and proinflammatory cytokine production. RedHill Biopharma (Yeliva/opaganib) ORAL
- **p38α/β inhibitor** — Reduces the pro-inflammatory cytokines such as IL-6 and corrects renin-angiotensin system dysfunction. Fulcrum Therapeutics (losmapimod) ORAL
- **MAP2K2 inhibitor** — Inhibits MK2-PI pathway and inhibits the production of proinflammatory cytokines. Aclaris Therapeutics (ATI-450) ORAL
- **PI3K-δ/γ inhibitor** — Repurposed leukemia drug which could inhibit the hyper-activation of innate immune system by polarizing macrophages. Secura Bio (Copiktra/duvelisib) ORAL
- **Balance arachidonic acid and docosahexaenoic acid (AA/DHA) levels** — Reduces pro-inflammatory cytokines and could interfere with coronavirus replication. Laurent Pharmaceuticals (LAU-7b/fenretinide) ORAL
- **TLR7/8 inhibitor** — Reduces the immune cell activation and inflammation by blocking two innate immune sensors. Merck KGaA (M5049) ORAL
- **RIPK1 inhibitor** — Inhibits TNF-α pathway. Denali Therapeutics/Sanofi (DNL-758/SAR443122) ORAL
- **Dual αVβ1/αVβ6 integrin inhibitor** — Blocks TGF-β activation. Pliant (PLN-74809) ORAL
- **Activate Vitamin D Receptor (VDR)** — VDR is highly expressed in lung alveolar type II cells (AcII) where it modulates anti-inflammatory effects and negatively regulates the renin-angiotensin pathway. (Vitamin D) ORAL
 - **Vitamin D receptor agonist**: Opko (Rayaldee/calcifediol ER) ORAL
- **NLRP3 inhibitor** — Prevents the hyperinflammation by inhibiting the production of IL-1β. Olatec Industries (dapansutrile) ORAL
- **IRAK4 inhibitor** — Inhibits TLR-induced inflammation and blocks the production of inflammatory cytokines in human monocytes. Pfizer (PF-06650833) ORAL
- **Melanocortin receptor-1/3 (MCR1/3) agonist** — Inhibits the release of pro-inflammatory cytokines from alveolar macrophages. SynAct Pharma AB (AP1189) ORAL
- **Glucokinase activator** — Activates the migration of Tregs to the sites of inflammation and reduces blood glucose to improve clinical outcomes in patients with diabetes. AstraZeneca (AZD1656) ORAL
- **Undisclosed mechanism** — Neuroprotective drug. NeuroActiva (NA-831) ORAL — *being tested +/-atazanavir and +/- dexamethasone

Less convenient delivery — IV or injection
- **Anti-CD6 antibody** — Repurposed psoriasis drug blocks ALCAM-mediated T-cell activation and downregulates IL-6, IL-2, and TNF-α.
 - **Approved for emergency use in India but based on limited RCT data (n=30, single country); requires larger global trial to support US and/or EU approval** — Itolizumab reduced mortality rate compared to best supportive care (BSC) arm (0% itolizumab vs. 30% BSC) in a small (n=30) RCT in India in which the BSC arm included steroids and antivirals. Equillium/Biocon (itolizumab) IV — *EUA in India; *decided not to initiate Ph3
- **CRAC inhibitor** — The CRAC channel is involved in the production of cytokines in T cells, and inhibiting it may lower levels of IL-6 and other inflammatory cytokines. CalciMedica (Auxora/CM-4620-IE) IV
- **Neuropilin 2 modulator** — Downregulates T-cell response, thereby reducing the production of inflammatory cytokines. aTyr (ATYR1923) IV
- **CD24-IgG1 Fc domain fusion protein I** — Dampens host response to tissue injury. CD24Fc in combination with SoC was shown to increase the likelihood of clinical recovery by 60% and to reduce the median recovery time compared to SoC (6 days vs 10 days). Merck/OncoImmune (Saccovid/CD24Fc) IV
- **Concentrate of human plasma-derived antibodies** — This product is meant to dampen an overactive immune response, unlike antibodies from patients that recovered from COVID-19, which are intended to target the virus. Octapharma (Octagam/Immune Globulin) IV; Grifols (Gamunex-C/immune globulin) IV; Biotest (Trimodulin) IV
- **HMGP1 inhibitor** — Heparin-derivative with anti-inflammatory properties. Chimerix (dociparstat) IV
- **Vasoactive intestinal peptide (VIP) receptor agonist** — Dampens inflammation via down regulating TNF-α, IFN-γ, IL-12, IL-17A, and IL-6. PhaseBio (PB1046) SUBQ
- **CD95L-Fc domain fusion protein** — Prevents apoptotic cell death of immune cells and epithelial cells. Apogenix (asunercept) IV
- **CTLA4 fusion peptide** — Repurposed rheumatoid arthritis drug that suppresses T-cell co-stimulation. Bristol-Myers Squibb (Orencia/abatacept) IV
- **TLR4 inhibitor** — Dampens the excessive inflammatory response and reduces IL-6, and IL-6 mediated acute lung injury. Edesa Biotech (EB05) IV — *Phase 3 is ongoing
- **Urinary trypsin inhibitor** — Repurposed sepsis drug down-regulates pro-inflammatory cytokines. (ulinastatin) IV
- **Anti-CD14 antibody** — Blocks the amplification of inflammatory response by CD14. Implicit Bioscience (IC14) IV
- **Low molecular weight fraction of human serum albumin (HSA)** — Repurposed knee osteoarthritis (OA) drug with Aryl Hydrocarbon Receptor (AhR) agonist properties which decreases levels of IL-23 and IL-17 and promotes the activation of anti-inflammatory macrophages. Ampio (Ampion) IV
- **Bradykinin B2 receptor inhibitor** — Repurposed approved hereditary angioedema (HAE) drug reduces inflammation and inhibits bradykinin-induced vasodilation. Takeda (Firazyr/icatibant*) SUBQ — *tested in combination with remdesivir
- **Recombinant plasma gelsolin** — Enhances macrophage nitric oxide synthase 3 (NOS3) function and enhances macrophage pathogen clearance. BioAegis (rhu-pGSN) IV
- **Substance P/neurokinin-1 (NK1) receptor antagonist** — Repurposed approved drug for prevention of nausea and vomiting after cancer chemotherapy, which decreases the production and release of pro-inflammatory cytokines. Heron (Cinvanti/aprepitant) IV
- **Radiolabelled urokinase plasminogen activator receptor (uPAR) antibody** — Delivers cytotoxic payload and kills selectively the aberrantly activated immune cells that express uPAR. Monopar Therapeutics/NorthStar Medical Radioisotopes (MNPR-101 radiolabelled) IV
- **Anti-ILT7 antibody** — Depletes plasmacytoid dendritic cells (pDCs) and decreases levels of TNF-α and IL-6. Horizon Therapeutics/Viela Bio (VIB7734) IV
- **Superoxide dismutase (SOD) mimetic** — Removing superoxide inhibits the hyperinflammatory cycle; thereby protecting lungs and other organs from damage. Galera (GC4419/avasopasem manganese) IV
- **L-citrulline replacement** — Increases plasma citrulline and arginine levels, restoring the homeostasis of nitric oxide synthase and reducing inflammation. Asklepion (L-Citrulline) IV
- **Anti-P-selectin antibody** — Decreases inflammation and prevents thrombosis by blocking leukocyte and platelet adherence to the vessel wall. Novartis (Adakveo/crizanlizumab) IV
- **Allosteric modulator of GABA-A receptor** — Repurposed postpartum depression drug decreases the release of pro-inflammatory cytokines. Sage Therapeutics (Zulresso/brexanolone) IV
- **Unclear mechanism** — Increases myeloid-derived suppressor cells and reduces inflammation. Shaperon (NuSepin) IV
- **Unclear mechanism**: Ashvattha Therapeutics (OP-101) IV/SUBQ

Improve lung function
- **Inhibit lung fibrosis**
 - **Multi-targeted TKI inhibitor (PDGFR, FGFR and VEGFR inhibitor)** — Repurposed drug with anti-fibrotic and anti-inflammatory effect currently approved for idiopathic pulmonary fibrosis (IPF) and systemic sclerosis associated interstitial lung disease (SSc-ILD). Boehringer Ingelheim (Ofev/nintedanib) ORAL
 - **Galectin-3 inhibitor** — Repurposed drug with anti-fibrotic and anti-inflammatory effect. Galecto Biotech (GB-0139) INHALED
 - **MMP-12 inhibitor** — Repurposed drug with anti-fibrotic and anti-inflammatory effect. Foresee Pharmaceuticals (FP-025) ORAL
- **Reduce pulmonary edema** — **Block CTGF**: Fibrogen (pamrevlumab) IV
- **Improve oxygen saturation** — Increasing vasodilation is likely to increase oxygen saturation.
 - **Repurposed CF drug** — Clear extracellular neutrophil traps (NETs). Roche (Pulmozyme/dornase alfa) INHALED
 - **Repurposed alpha-1 antitrypsin deficiency (AATD) drug** — Neutrophil elastase inhibitor inhibits NET formation and NET activity.
 - **Neutrophil elastase inhibitor** — Inhibits NET formation and NET activity. Mereo (alvelestat) ORAL
 - **Alpha-1 proteinase inhibitor** — Protects the airway from damage by preventing excessive accumulation of activated neutrophil elastase. Grifols (alpha-1 proteinase inhibitor) IV
 - **Repurposed erectile dysfunction drugs** — Vasoactive intestinal peptide (VIP) binds to VIP receptor (VIPAC1) on alveolar type II cells in the lung, up regulates surfactant production, blocks apoptosis and blocks the production of TNF-α. VIP also inhibits SARS-CoV-2 replication in vitro. NeuroRx/Relief Therapeutics (RLF-100/aviptadil) IV/INHALED — *Phase on top of remdesivir
 - **Also potential for antiviral activity: nitric oxide** — Nitric oxide therapy may additionally inactivate the virus by creating free radicals. Mallinckrodt (INOmax) INHALED; Thirty Respiratory (RESP301) INHALED; Bellerophon Therapeutics (INOpulse) INHALED
 - **Repurposed COPD drug**: (almitrine) IV
 - **Repurposed PAH drug**: Actelion (Ventavis/iloprost) IV
 - **Re-oxygenates hypoxic tissue**: Diffusion Pharmaceuticals (trans sodium crocetinate) IV
 - **Hypoxia-inducible factor prolyl hydroxylase (HIF-PH) inhibitor**: Akebia Therapeutics (vadadustat) ORAL
 - **Repurposed RDS drug**: Chiesi (Curosurf/poractant alfa) BRONCHIAL FIBROSCOPY
 - **Undisclosed mechanism**: Merck (MK-5475) INHALED
- **Undisclosed mechanism**: Novartis (MAS825) IV
- **Restore glutathione activity** — Repurposed rheumatoid arthritis drug restores glutathione activity and inhibits reactive oxygen species (ROS) generation that promote acute lung injury. ReviveMed (bucillamine/REV-002) ORAL
- **Promote lung tissue regeneration and prevent lung fibrosis** — Hepatocyte growth factor (HGF) agonist. Angion (ANG-3777) IV
- **Reduce airway injury** — TRPC6 channel blocker reduces the effects of reactive oxygen species, preventing swelling of the lung tissue. Boehringer Ingelheim (BI 764198) ORAL

Target lung angiogenesis — Beyond cancer, angiogenesis known to be dysregulated in both chronic lung disease and inflammatory conditions.
- **Reduce angiopoietin-2 (Ang2)** — Angiopoietin-2 promotes angiogenesis and inflammation in the presence of VEGF; anti-Ang2 mAbs inhibit VEGF-mediated signaling. Eli Lilly (LY3127804) IV
- **Inhibit vascular endothelial protein tyrosine phosphatase (VE-PTP)** — Activate Tie2 signaling to stabilize blood vessels and maintain vascular integrity and overcome the effects of Ang-2 signaling. Aerpio Pharmaceuticals (razuprotafib) ORAL

Prevent organ failure beyond the lung

Some patients infected with COVID-19 have experienced rapid onset of abnormal blood clotting, which can lead to stroke.

Treat abnormal clotting
- **Anticoagulants** — Agents that block synthesis or function of blood clotting factors.
 - **Easier to administer: oral or subcutaneous injection**
 - **Factor Xa inhibitor**: Bayer/J&J (Xarelto/rivaroxaban) ORAL; (fondaparinux) SUBQ
 - **Low molecular weight heparin**: (enoxaparin) SUBQ; Pfizer (Fragmin/dalteparin) SUBQ — *Ex-US
 - **TF inhibitor**: ARCA Biopharma (AB201/rNAPc2) SUBQ
 - **Repurposed blood thinner drug**: (apixaban) ORAL; (aspirin) ORAL
 - **Harder to administer: IV infusion**: (heparin) IV
 - **Factor XIIa antibody**: CSL Behring (CSL312/garadacimab) IV
 - **Plasma kallikrein inhibitor**: Takeda (Takhzyro/lanadelumab) IV
- **Tissue plasminogen activator (tPA) that breaks down clots** — tPA is a serine protease that breaks the bonds between proteins that make up blood clots. Roche (Activase/alteplase) IV; Takeda (Actilyse/alteplase) IV
- **Repurposed drug previously approved for stroke prevention**: Boehringer Ingelheim (Aggrenox/aspirin-dipyridamole) ORAL

Protect organs from additional damage
- **Protect heart and kidney from damage** — Inhibit sodium-glucose transport to prevent glucose-reabsorption in the kidney and improve cardiac function by lowering hemodynamic stress. AstraZeneca (Farxiga/dapagliflozin) ORAL
- **Protect liver and kidney from damage** — Sulfated oxysterol stabilizes mitochondria, reduces lipotoxicity, regulates inflammatory responses and promotes cell survival. Durect (DUR-928) IV
- **Protect kidney from additional damage** — **Eliminate reactive oxygen species (ROS) and reactive nitrogen species (RNS) to reduce kidney damage**: Renibus Therapeutics (RBT-9) IV
- **Protect heart from additional damage**
 - **Inhibit aldose reductase to prevent cardiomyopathy**: Applied Therapeutics (AT-001) ORAL
 - **Repurposed drug previously approved to reduce risk of cardiovascular events in patients with elevated triglyceride levels**: Amarin (Vascepa/icosapent ethyl) ORAL

COVID-19 THERAPEUTICS: TARGET LUNG INFLAMMATION + ARDS MILESTONES* (DATA AS OF FEBRUARY 11, 2022)

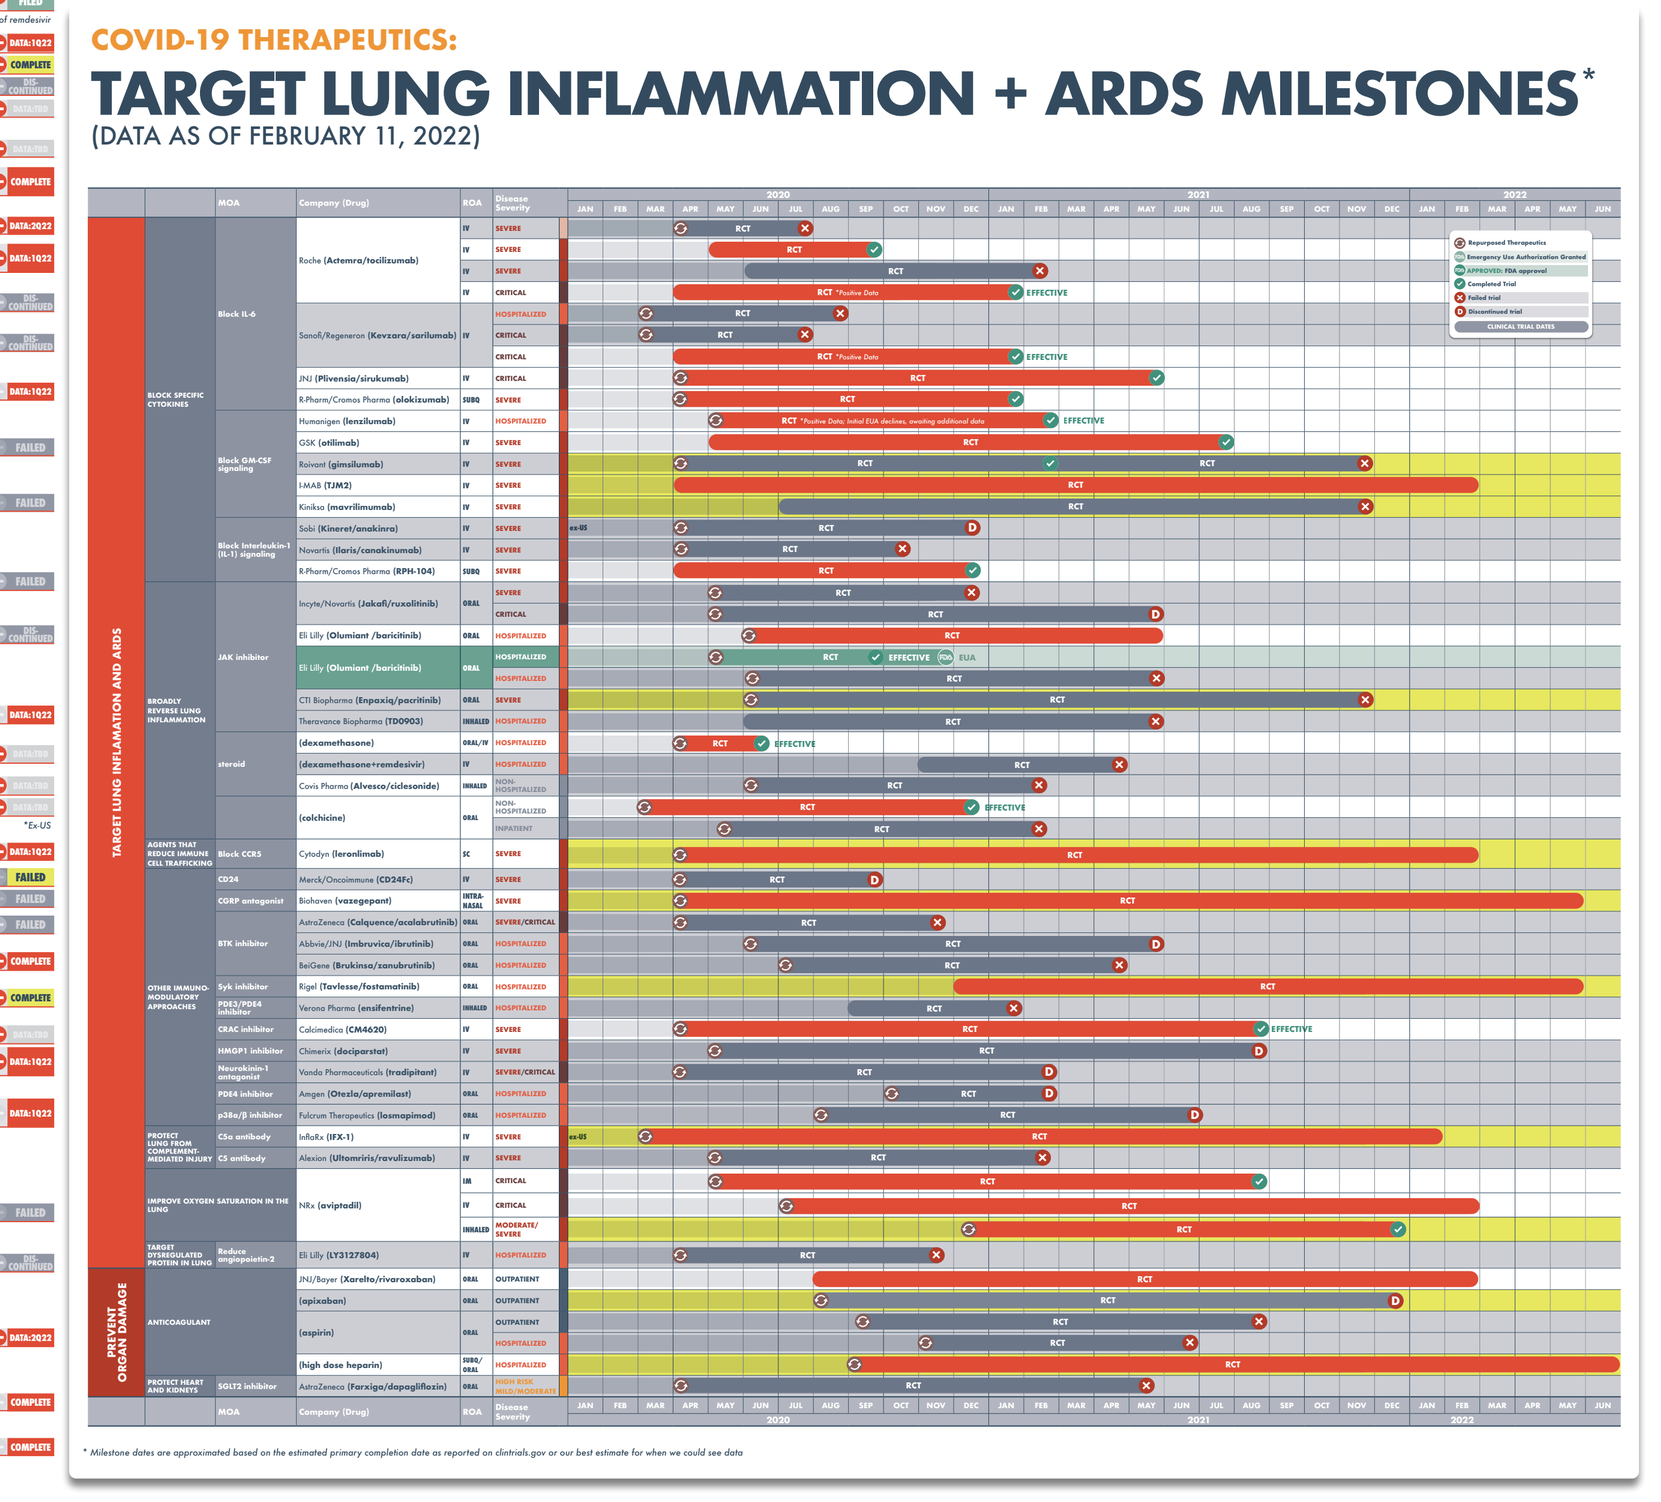

* Milestone dates are approximated based on the estimated primary completion date as reported on clinicaltrials.gov or our best estimate for when we could see data